318a ASCO ANNUAL MEETING, JUNE 1-5, 2018 | McCORMICK PLACE, CHICAGO, IL, USA

Phase 2 Study to Assess the Efficacy and Safety of Autologous Tumor Infiltrating Lymphocytes (LN-145) Alone and In Combination with Anti-PD-L1 Inhibitor Durvalumab (MEDI4736) in Patients with Locally Advanced or Metastatic Non-Small Cell Lung Cancer (NSCLC)

Sylvia Lee[1], Liza Villaruz[2], Susie Tanamly[3], Igor Gorbatchevsky[3], Corina Andresen[3], Sam Suzuki[3], Maria Fardis[3], Missak Haigentz[4]

[1]University of Washington, Seattle, WA; [2]University of Pittsburgh Medical Center - Hillman Cancer Center, Pittsburgh, PA; [3]Iovance Biotherapeutics, San Carlos, CA; [4]Atlantic Health System/Morristown Medical Center, Morristown, NJ

MedImmune

IOVANCE BIOTHERAPEUTICS

ADVANCING IMMUNO-ONCOLOGY

999 Skyway Road, STE 150, San Carlos, CA 94070

For more information, please contact
leesm@uw.edu
clinical.inquiries@iovance.com

BACKGROUND

- Lung cancer is the leading cause of human cancer deaths worldwide, with approximately 1.7 million deaths reported in 2015, of which 80% to 85% were attributed to non-small cell lung cancer (NSCLC).[1]
- For patients with locally advanced or metastatic disease, standard chemotherapy shows objective response rate (ORR) of 10% to 40%,[2,3] and a median survival of approximately 1 year.[4,5]
- Adoptive cell therapy (ACT) with tumor infiltrating lymphocytes (TIL) has demonstrated the potential for durable complete responses in immunogenic tumors with high mutational burden, such as melanoma, in studies conducted at National Cancer Institute (NCI).[6]
- A positive correlation has been demonstrated between the presence of TIL in NSCLC tumor specimens and patient outcome.[7,8]
- Recently, the anti-PD-L1 monoclonal antibody durvalumab (Imfinzi®) was approved for patients with locally advanced, unresectable Stage III NSCLC whose disease has not progressed following chemoradiotherapy.[9]
- Durvalumab dosing prior to harvest of tumor for LN-145 generation is expected to promote increased TIL trafficking into tumor lesions. In addition, anti-PD-L1 treatment prior to and immediately following TIL (LN-145) infusion may further dampen a suppressive tumor microenvironment, allowing improved engraftment and potency of the infused TIL (LN-145).[10,11]
- This study was designed to evaluate the efficacy and safety of LN-145 (an autologous investigational TIL therapy) given alone or in combination with durvalumab for the treatment of patients with Stage III or Stage IV NSCLC who are checkpoint therapy-naïve and who have received ≥ 1 line of prior systemic therapy in the locally advanced or metastatic setting.

Figure 1. Iovance cryopreserved LN-145 manufacturing process (22 days)

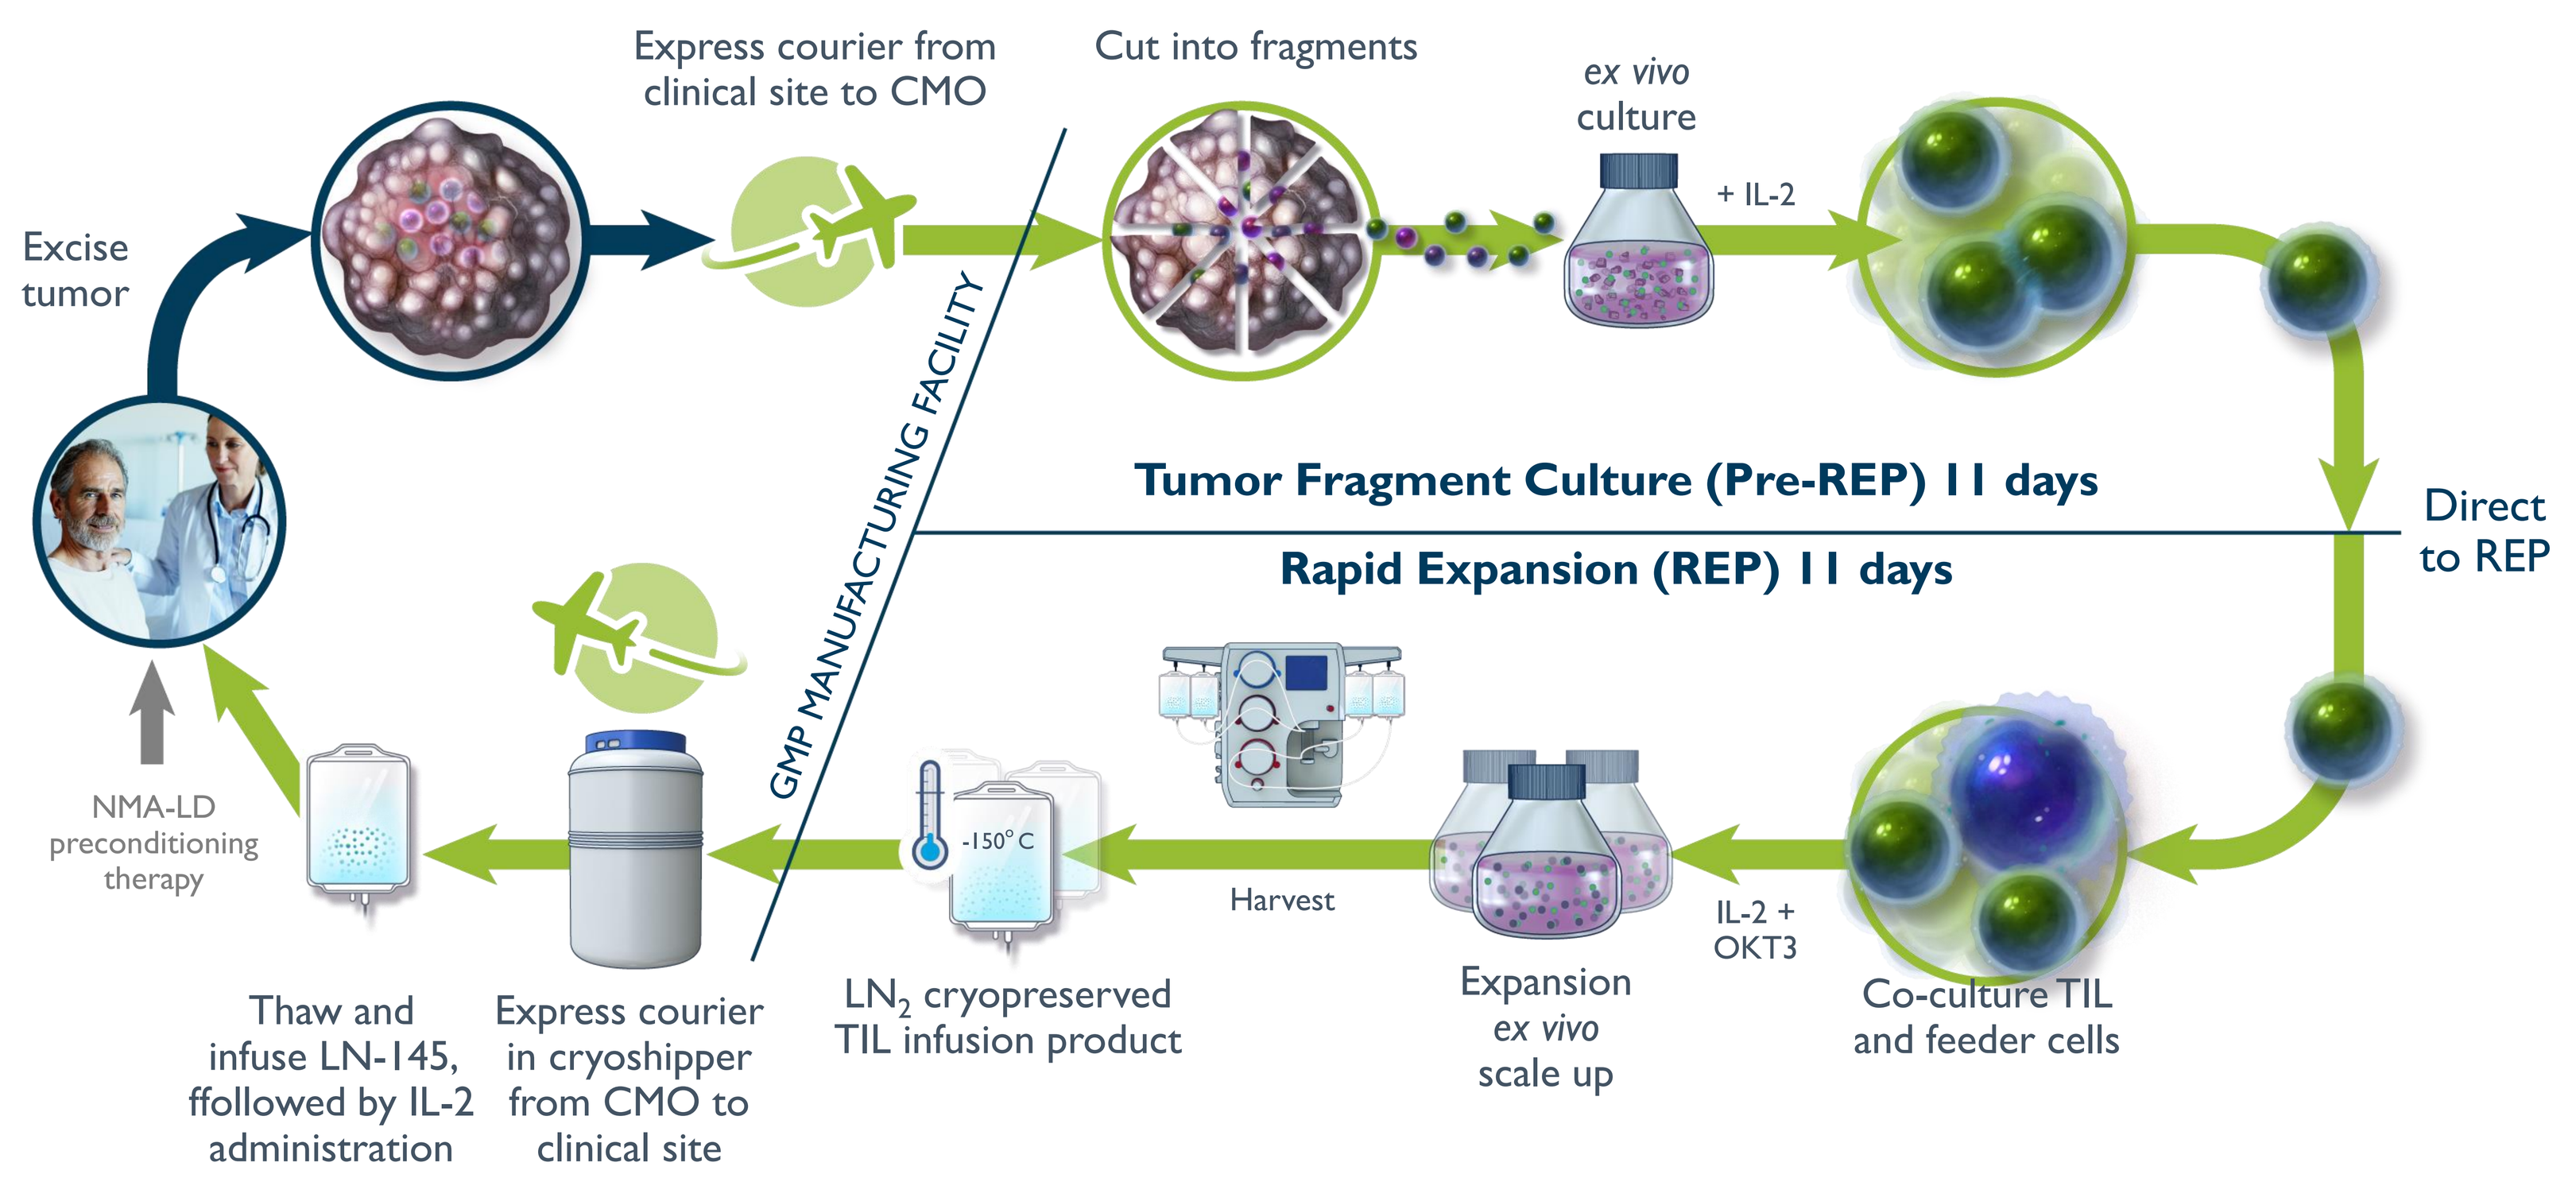

Excise tumor
Express courier from clinical site to CMO
Cut into fragments
ex vivo culture
+ IL-2
GMP MANUFACTURING FACILITY
Tumor Fragment Culture (Pre-REP) 11 days
Rapid Expansion (REP) 11 days
Direct to REP
Co-culture TIL and feeder cells
IL-2 + OKT3
Expansion ex vivo scale up
Harvest
-150° C
LN2 cryopreserved TIL infusion product
Express courier in cryoshipper from CMO to clinical site
Thaw and infuse LN-145, ffollowed by IL-2 administration
NMA-LD preconditioning therapy

STUDY OVERVIEW

- Phase 2, multicenter, open-label, 2-cohort study evaluating ACT with autologous TIL therapy (LN-145) alone in Cohort 1, or in combination with durvalumab in Cohort 2
- Approximately 10 investigational centers in the US
- The planned sample size is 24 treated patients; 12 patients in each cohort
- The primary statistical analysis is based on the use of descriptive methods and estimation of efficacy and safety parameters performed by treatment cohort

NCT03419559

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster

STUDY DESIGN

- LN-145 is prepared at a central GMP facility from TIL extracted from surgically-resected tumors. LN-145 infusion is preceded by a non-myeloablative lymphodepletion regimen of cyclophosphamide (60 mg/kg x 2 days) and fludarabine (25 mg/m^2 x 5 days), followed by up to 6 intravenous infusions of IL-2 (600,000 IU/kg)
- Cohort 1 patients receive LN-145 therapy alone
- Cohort 2 patients receive 1500 mg IV durvalumab as follows: 2 weeks prior to and 2 weeks after tumor harvest; then 2 weeks following LN-145 infusion and continue 1500 mg IV durvalumab Q4W until disease progression or unacceptable toxicity
- Patients in either cohort unable to receive LN-145 are allowed to go on to receive durvalumab alone

Figure 2. Study Design

Now enrolling at sites in US

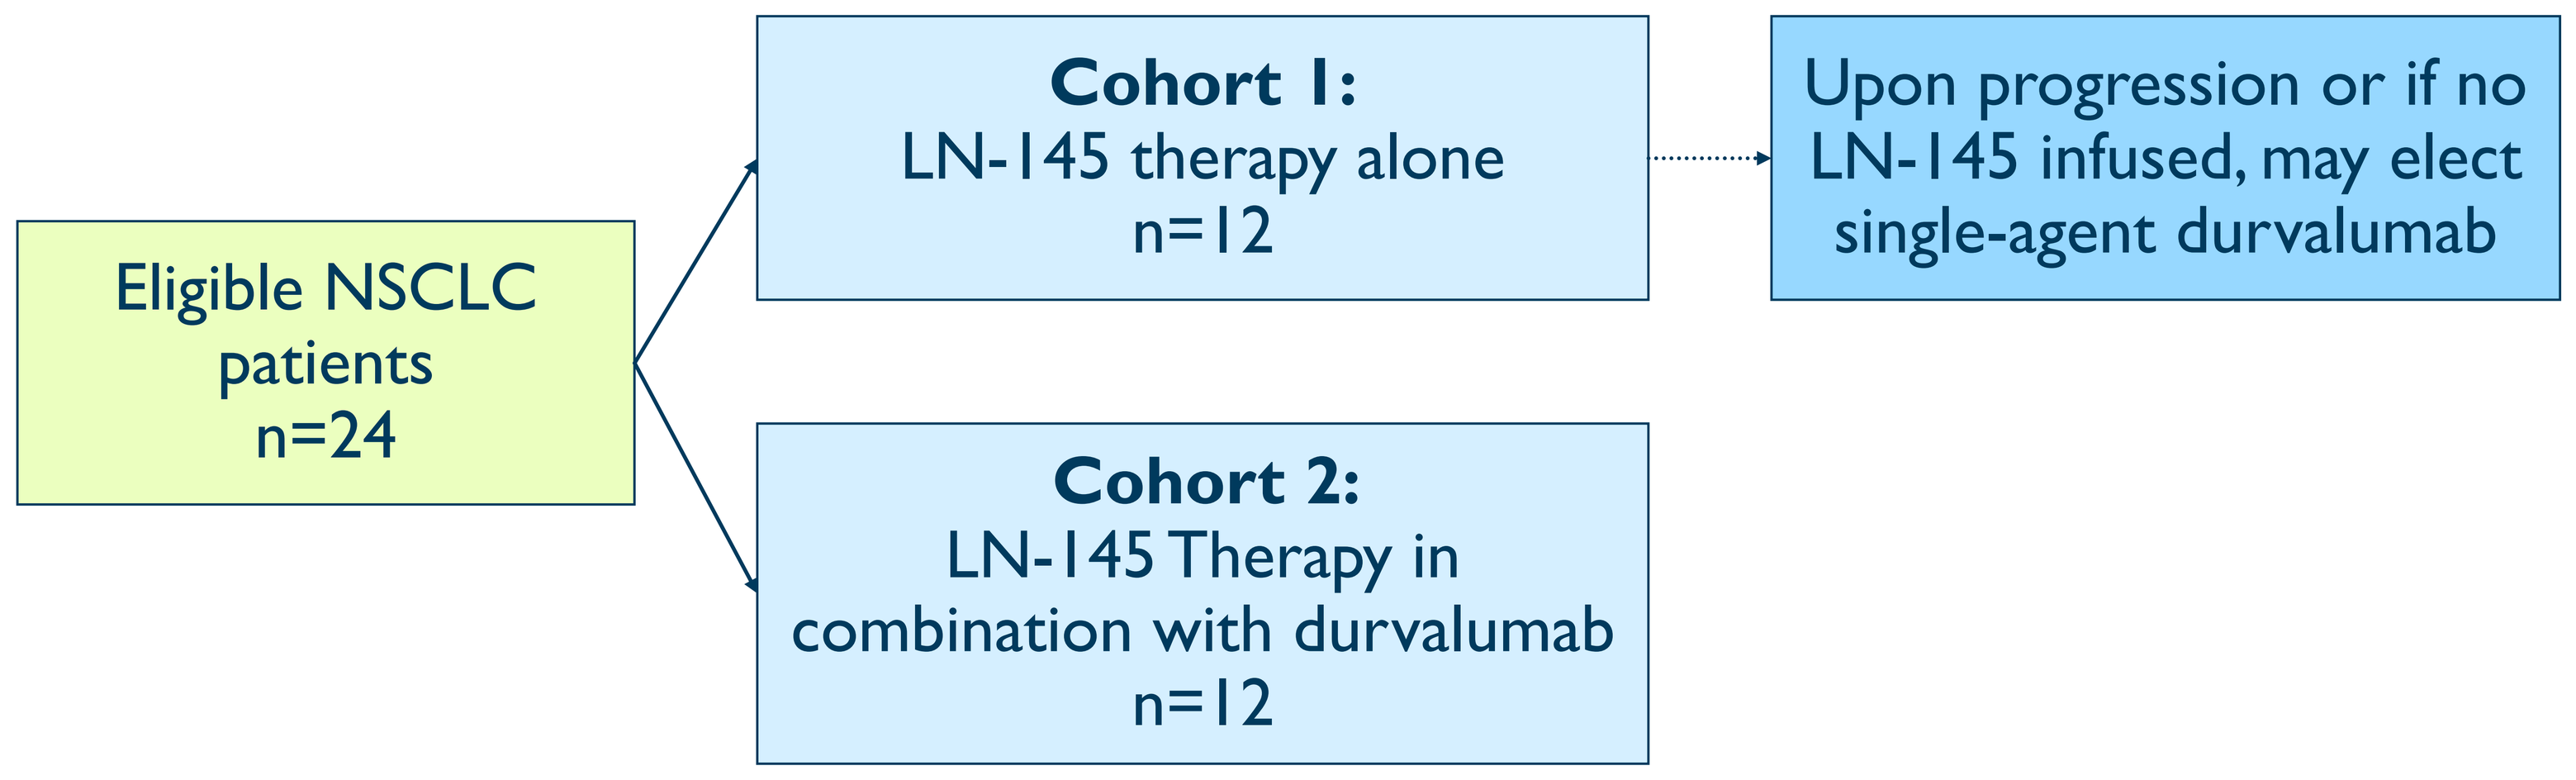

Eligible NSCLC patients n=24
Cohort 1: LN-145 therapy alone n=12
Upon progression or if no LN-145 infused, may elect single-agent durvalumab
Cohort 2: LN-145 Therapy in combination with durvalumab n=12

OUTCOME MEASURES

Primary:
- Objective response rate (ORR)
- Safety evaluation

Secondary:
- Duration of response (DOR), progression-free survival (PFS), and overall survival (OS)

Exploratory:
- Complete response (CR) rate and disease control rate (DCR)
- Persistence of LN-145 and immune correlates of response, survival, and toxicity of the treatment
- Efficacy per irRECIST[12]
- Health-related quality of life (HRQoL)

MAJOR INCLUSION & EXCLUSION CRITERIA

Major Inclusion Criteria
- Histologically/cytologically confirmed diagnosis of Stage III or Stage IV NSCLC (squamous, nonsquamous, adenocarcinoma, large cell carcinoma), and have received ≥ 1 line of prior systemic therapy in the locally advanced or metastatic setting;
- One tumor lesion resectable for TIL generation and ≥ one tumor lesion for RECIST assessment as target;
- Male or female, 18 years of age or older;
- Minimum of 21 days or 5 half-lives washout from last dose of tumor-directed therapy to the first study treatment, whichever is longer period to the first study treatment;
- ECOG performance status of 0 or 1, and estimated life expectancy of ≥3 months;
- Adequate bone marrow, liver, and renal function;
- Seronegative for the HIV Ab, Hepatitis B Ag and Hepatitis C Ab or Ag.

Major Exclusion Criteria
- Prior cell transfer therapy;
- Prior anti-PD-1 or anti-PD-L1 inhibitors (including durvalumab); other prior immunotherapy(ies) allowed;
- Active or prior documented autoimmune or inflammatory disorders or active infections;
- History of primary immunodeficiency, history of allogeneic organ transplant that requires therapeutic immunosuppression;
- History of hypersensitivity to any components of TIL therapy, and other study drugs: cyclophosphamide, fludarabine, or IL-2;
- LVEF < 45%;
- FEV_1 ≤ 60%;
- Active central nervous system metastases and/or leptomeningeal disease;
- History of other malignancies, except for curatively treated with no evidence of disease for ≥ 3 years.

SUMMARY

- Locally advanced or metastatic NSCLC presents a high unmet medical need with low survival rates and limited effective treatment options.
- The presence of TIL in tumor lesions has been correlated with improved outcomes in a number of solid tumors, including NSCLC.[7,8]
- Our hypothesis is that the combination of the anti-PD-L1 checkpoint inhibitor durvalumab and LN-145 therapy may further enhance the efficacy and persistence of TIL.
- This study aims to assess the potential of TIL therapy with LN-145 either alone or in combination with durvalumab for the treatment of patients with locally advanced or metastatic NSCLC.

Disclosure
This study and poster are sponsored by Iovance Biotherapeutics, Inc., in collaboration with MedImmune, the global biologics research and development arm of AstraZeneca

References

1. Siegel RL, Miller KD, Jemal A. Cancer statistics, 2015. *CA Cancer J Clin*. 2015;65(1):5-29.
2. Molina JR, Yang P, Cassivi SD, Schild SE, Adjei AA. Non-small cell lung cancer: epidemiology, risk factors, treatment, and survivorship. *Mayo Clin Proc*. 2008;83(5):584-94.
3. Bonomi PD. Implications of key trials in advanced non-small cell lung cancer. *Cancer*. 2010;116(5):1155-64.
4. Leighl NB. Treatment paradigms for patients with metastatic non-small-cell lung cancer: first-, second-, and third-line. *Curr Oncol*. 2012;19(Suppl 1):S52-8.
5. Gerber DE, Schiller JH. Maintenance chemotherapy for advanced non-small-cell lung cancer: new life for an old idea. *J Clin Oncol*. 2013;31(8):1009-20.
6. Goff SL, Dudley ME, Citrin DE, Somerville RP, Wunderlich JR, Danforth DN, et al. Randomized, Prospective Evaluation Comparing Intensity of Lymphodepletion Before Adoptive Transfer of Tumor-Infiltrating Lymphocytes for Patients With Metastatic Melanoma. *J Clin Oncol*. 2016;34(20):2389-97.
7. Schalper KA, Brown J, Carvajal-Hausdorf D, McLaughlin J, Velcheti V, Syrigos KN, et al. Objective measurement and clinical significance of TILs in non-small cell lung cancer. *J Natl Cancer Inst*. 2015;107(3).
8. Reynders K, De Ruysscher D. Tumor infiltrating lymphocytes in lung cancer: a new prognostic parameter. *J Thorac Dis*. 2016;8(8):E833-5.
9. AstraZeneca Imfinzi (durvalumab) injection for intravenous use prescribing information 2018 [Available from: *https://www.azpicentral.com/imfinzi/imfinzi.pdf (accessed March 28, 2018)*].
10. Juneja VR, McGuire KA, Manguso RT, LaFleur MW, Collins N, Haining WN, et al. PD-L1 on tumor cells is sufficient for immune evasion in immunogenic tumors and inhibits CD8 T cell cytotoxicity. *J Exp Med*. 2017;214(4):895-904.
11. Spranger S, Spaapen RM, Zha Y, Williams J, Meng Y, Ha TT, et al. Up-regulation of PD-L1, IDO, and T(regs) in the melanoma tumor microenvironment is driven by CD8(+) T cells. *Sci Transl Med*. 2013;5(200):200ra116.
12. Bohnsack O, Ludajic K, Hoos A. Adaptation of the Immune-related Response Criteria: irRECIST. ESMO2014.

© 2018, Iovance Biotherapeutics