Real-World, Evidence-Based, Retrospective Study of Patients Infused With Commercially Released Lifileucel for Advanced Melanoma

Lilit Karapetyan, MD, MS, FACP,[1] Justin Moser, MD,[2] Barbara Ting-Wen Ma, MD, MS,[3] Christopher Otteni, MD, MBA,[1] Elizabeth Cathcart, BS,[4] Kimberly Loo, MD,[3] Jay Jacoby, BA,[2] Jamie K. Chau,[1] James W. Smithy, MD, MHS,[4] Nikhil Nayee,[2] Anna C. Pavlick, DO,[3] Ahmed Tawashi,[5] Amritha Seshaadri,[5] Min Yi, PhD,[5] Friedrich Graf Finckenstein, MD, PhD,[5] Brian Gastman, MD,[5] and Alexander N. Shoushtari, MD[4]

[1]H. Lee Moffitt Cancer Center & Research Institute, Tampa, FL; [2]HonorHealth Research Institute and Arizona State University John Shufeldt School of Medicine and Medical Engineering, Scottsdale, AZ; [3]Weill Cornell Medicine, New York, NY; [4]Memorial Sloan Kettering Cancer Center, Weill Cornell Medical College, New York, NY; and [5]Iovance Biotherapeutics, San Carlos, CA

Objective

- Report the first, multicenter, retrospective study of standard-of-care lifileucel TIL therapy in patients with metastatic melanoma in the real-world setting

Conclusions

- Lifileucel demonstrated meaningful real-world clinical activity in patients with advanced melanoma, confirming its clinical benefit as standard-of-care therapy
- Higher response rates were observed in patients with lower disease burden and fewer prior lines of therapy, supporting earlier referral for lifileucel
- Clinical benefit was independent of infused TIL dose
- Responders received fewer IL-2 doses, supporting that efficacy is not driven by IL-2 exposure
- No new safety signals or early mortality were observed

Background

- There is increasing interest in evaluating the real-world clinical activity of standard-of-care (SOC) lifileucel, particularly given potential differences in patient populations arising from the interval between the pivotal trial and subsequent regulatory approval; the real-world effectiveness of commercially available lifileucel has not yet been characterized
- Due to centralized, scalable manufacturing, the tumor-infiltrating lymphocyte (TIL) therapy lifileucel has emerged as a subsequent, SOC therapy for patients with metastatic melanoma capable of achieving durable remissions[1-4]
 - In the C-144-01 phase 2 study of lifileucel, a 31% objective response rate (ORR) and ~20% 5-year overall survival (OS) was observed[3]
 - Lifileucel received accelerated approval from the US FDA in Feb 2024, becoming the first approved cell therapy for a solid tumor[4]
- First-line immune checkpoint inhibitors (ICI) have transformed survival outcomes for patients with metastatic melanoma; however, responses to subsequent systemic therapies and resistance to ICI limit OS[5-11]
 - Patients harboring a *BRAF* V600 mutation who have been treated with BRAF/MEK inhibitors achieve high initial responses, but responses are often not durable[12,13]

Methods

Figure 1. Treatment Schema

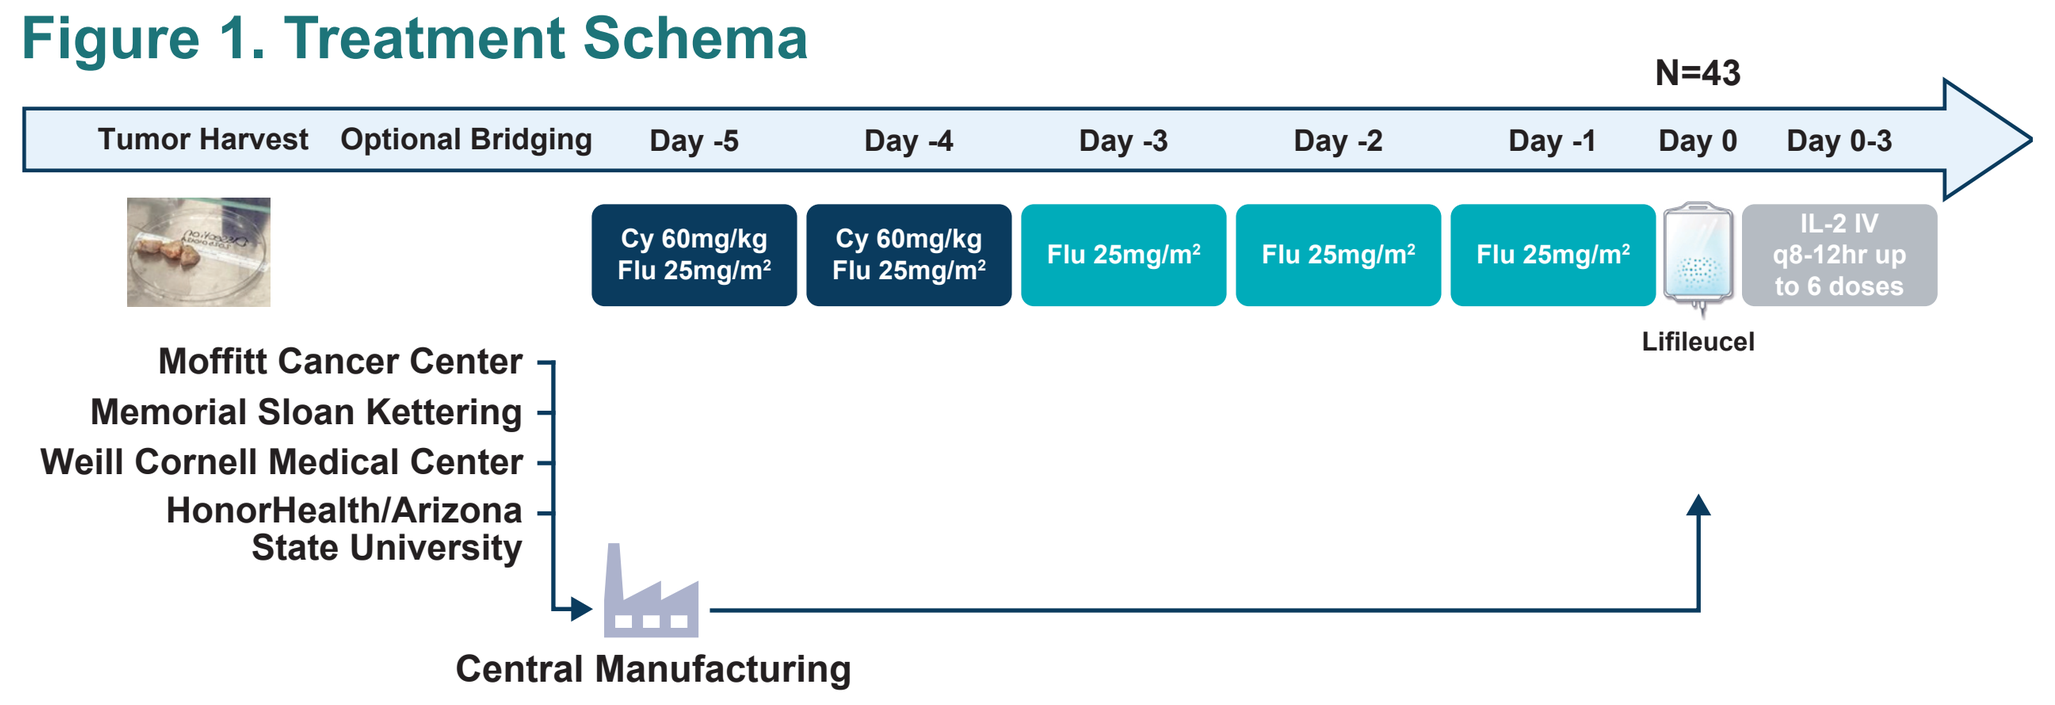

N=43
Tumor Harvest
Optional Bridging
Day -5
Day -4
Day -3
Day -2
Day -1
Day 0
Day 0-3
Cy 60mg/kg Flu 25mg/m²
Cy 60mg/kg Flu 25mg/m²
Flu 25mg/m²
Flu 25mg/m²
Flu 25mg/m²
IL-2 IV q8-12hr up to 6 doses
Lifileucel
Moffitt Cancer Center
Memorial Sloan Kettering
Weill Cornell Medical Center
HonorHealth/Arizona State University
Central Manufacturing

Dose and schedule modifications of lymphodepletion were allowed according to institutional practices. Patient selection varied between participating centers; Moffitt Cancer Center and Weill Cornell Medical Center included all patients treated with standard-of-care lifileucel, and Memorial Sloan Kettering and HonorHealth/Arizona State University selected patients who were primary refractory to anti-PD-1 therapy.

- Eligible patients were ≥18 years with advanced melanoma and previously received ICI therapy and, if harboring *BRAF* V600 mutation, BRAF/MEK inhibitors as well as ≥2 radiographic and/or clinical response assessments
- The study period occurred from May 16, 2024 through May 13, 2025 across four study sites where tumors were procured for centralized manufacturing (**Figure 1**)
- Cyclophosphamide (Cy; 60 mg/kg on days -5 and -4) and fludarabine (Flu; 25 mg/m^2 on days -5 to -1) were administered for lymphodepletion prior to lifileucel infusion (**Figure 1**)
 - Interleukin-2 (IL-2, 600,000 IU/kg) was administered every 8-12 hours after lifileucel infusion for up to six doses
- The primary study endpoint was physician-assessed ORR
 - Secondary endpoints were progression-free survival (PFS) and OS; safety assessments included preliminary evaluation by voluntary adverse event (AE) reporting
 - Data collected through Oct 2025 are included in this analysis

Results

Table 1. Baseline Patient Disease Characteristics

Characteristic	N=43
Median age, years (range)	59 (28–79)
Sex, n (%)	
Male	23 (54)
Female	20 (46)
Melanoma subtypes, n (%)	
Non-acral cutaneous	30 (70)
Acral	4 (9)
Mucosal	8 (19)
Unknown, primary	1 (2)
Distant metastasis (M stage), n (%)	
M1a	9 (21)
M1b	5 (12)
M1c	17 (40)
M1d	12 (28)
Liver metastases, n (%)	13 (30)
Brain metastases, n (%)	12 (28)
LDH prior to lymphodepletion, n (%)	
≤ULN	17 (40)
1–2× ULN	21 (49)
>2× ULN	5 (12)
BRAF V600 mutation, n (%)	19 (44)
Median number of prior systemic therapies (range)	3 (1–7)

- A total of 43 patients with metastatic melanoma who received lifileucel were included in this analysis (**Table 1**)

Table 2. Tumor Harvest Sites and Treatment Administration

Characteristic	N=43
Tumor harvest site, n (%)[a]	
Lymph node	17 (40)
Subcutaneous	11 (26)
Soft tissue	9 (21)
Lung	5 (12)
Liver	4 (9)
Spleen	1 (2)
Median time from harvest to lymphodepletion, days (range)	44 (35–93)
Bridging therapy, n (%)	19 (44)
Median number of infused TILs (range)	26×10^9 (11–53)
Median number of IL-2 doses (range)	5 (1–6)

[a]Multiple tumor harvest anatomic sites may be present in a patient.

- Tumors were most often harvested from lymph nodes (40%) and subcutaneous tissue (19%; **Table 2**)
 - Patients received a median of 26×10^9 infused TILs
 - All patients received at least 1 dose of IL-2, with a median of 5 doses

Figure 2. Clinical Outcomes Following Lifileucel Treatment

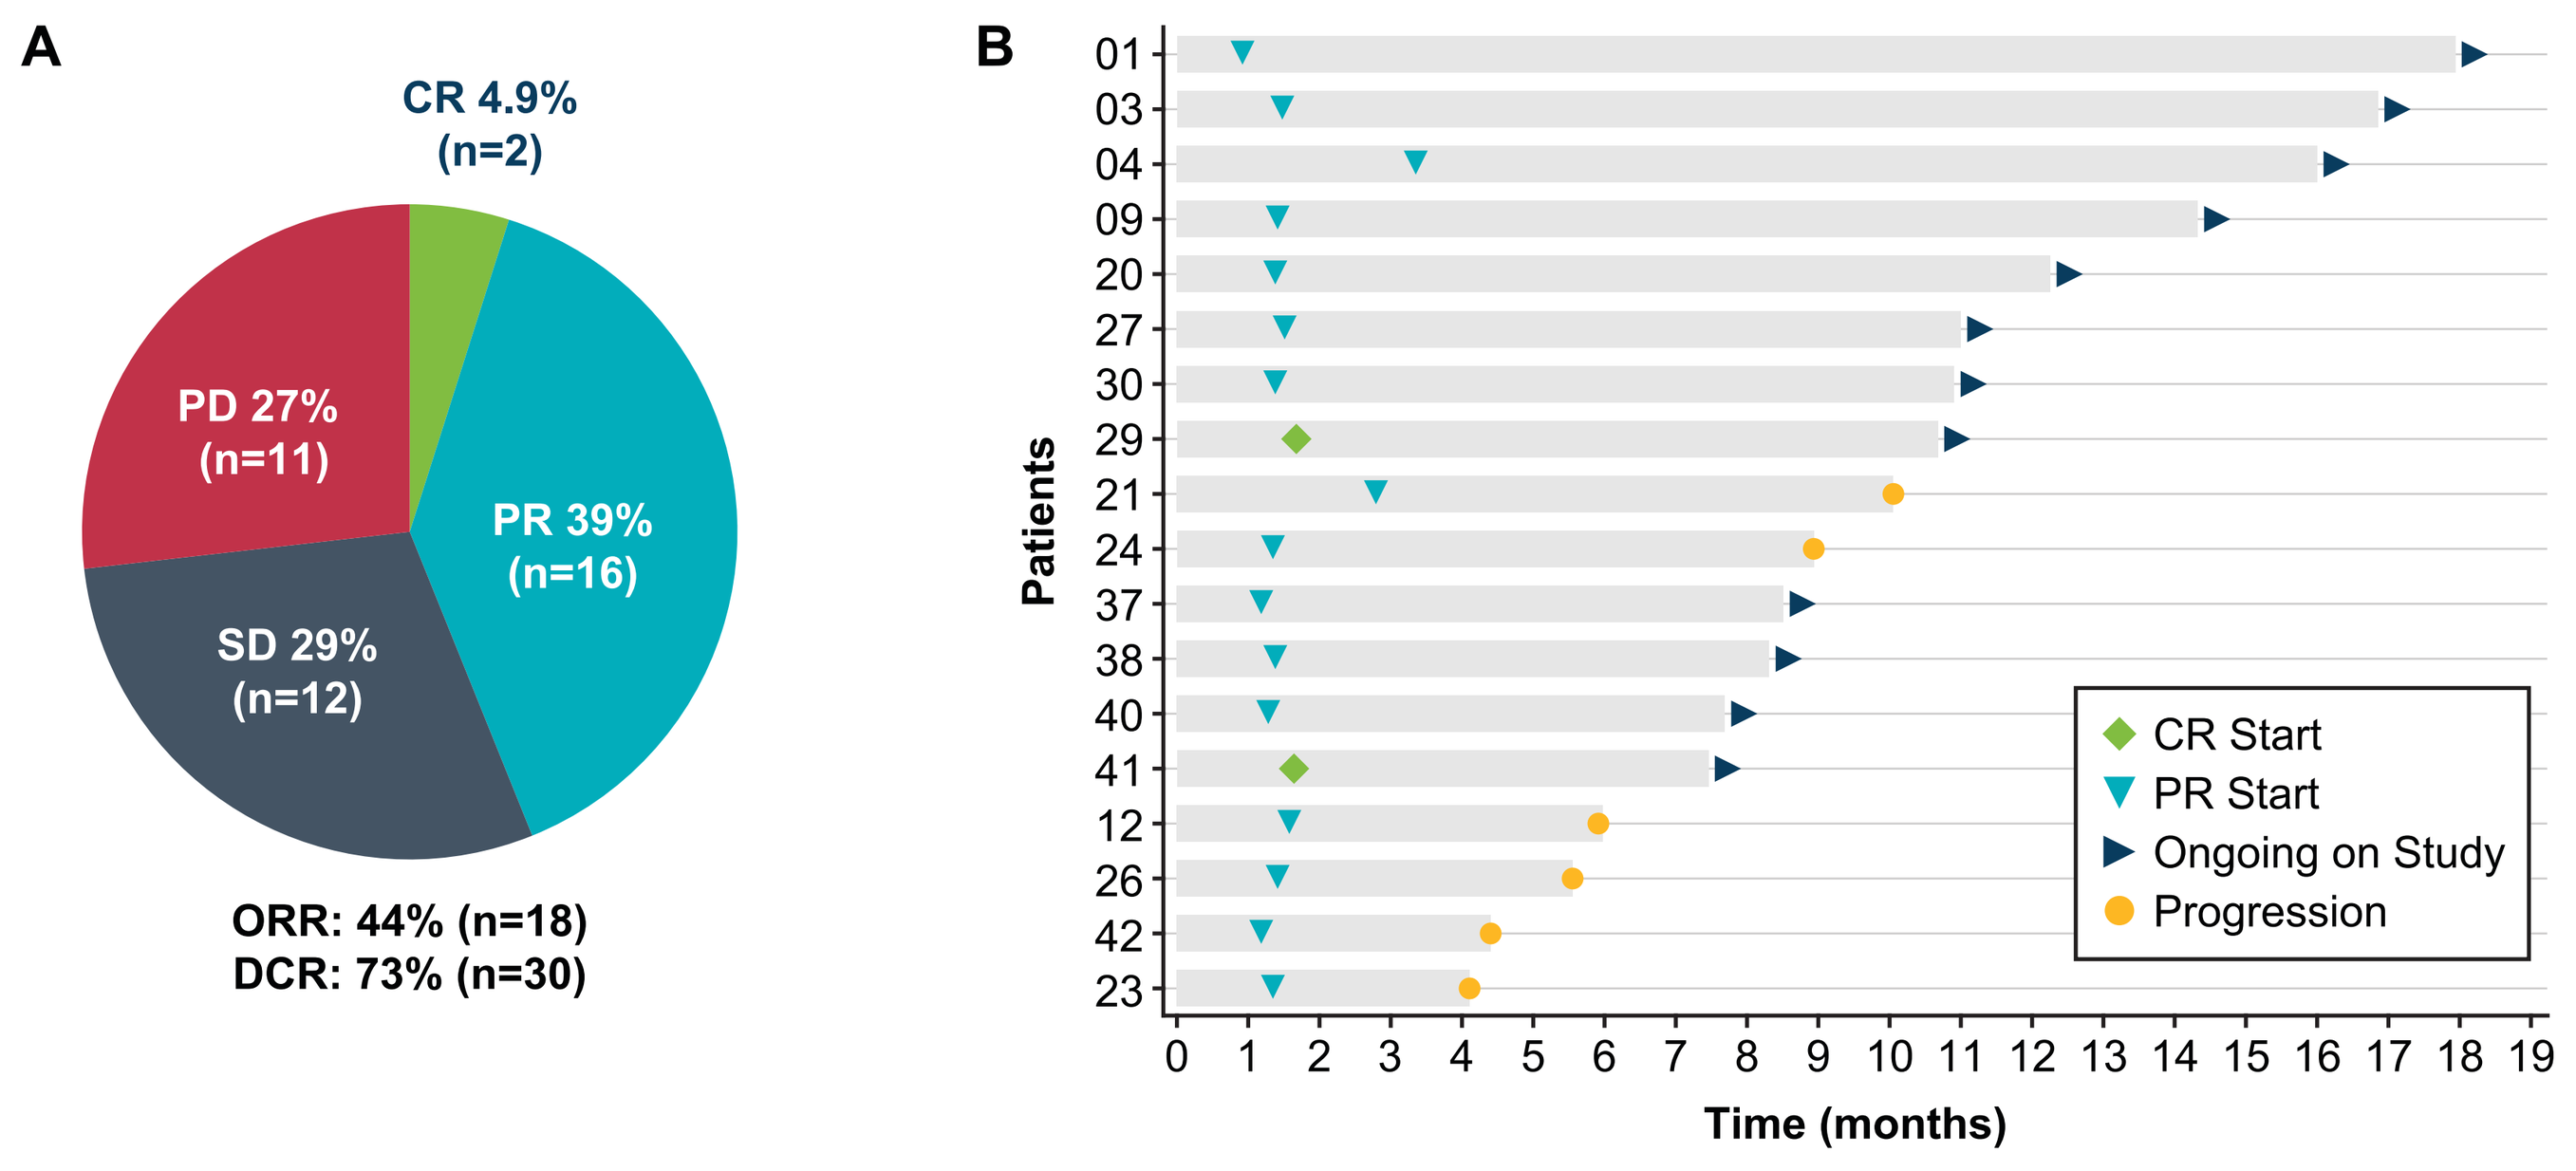

A
CR 4.9% (n=2)
PR 39% (n=16)
SD 29% (n=12)
PD 27% (n=11)
ORR: 44% (n=18)
DCR: 73% (n=30)
B
Patients
01 03 04 09 20 27 30 29 21 24 37 38 40 41 12 26 42 23
CR Start
PR Start
Ongoing on Study
Progression
Time (months)

(A) Best overall response to lifileucel therapy. The pie chart shows the distribution of CR (5%), PR (39%), SD (29%), and PD (27%). ORR was 44%, and the DCR was 73%. Out of 43 patients, 2 did not have measurable disease after tumor harvest; ORR was calculated based on patients with measurable lesions after tumor harvest (n=41). **(B)** Swimmer plot showing time from TIL infusion to first response and end of follow-up for individual patients. Each bar represents the time from TIL infusion up to date of new anti-cancer therapy, end of assessment, death, or data cutoff date, whichever occurs earlier. Symbols on the bars indicate the onset of responses (CR or PR), the end of response (progression) and follow-up status.

- Median follow up time was 6 months
- The ORR was 44%, with 5% having a complete response (CR) and 39% having a partial response (PR; **Figure 2A**)
- The disease control rate was 73% (**Figure 2A**)
- Most responses were observed at the first radiologic assessment, with 12 ongoing during follow-up (**Figure 2B**)

Figure 3. Subgroup Analyses Following Lifileucel Treatment

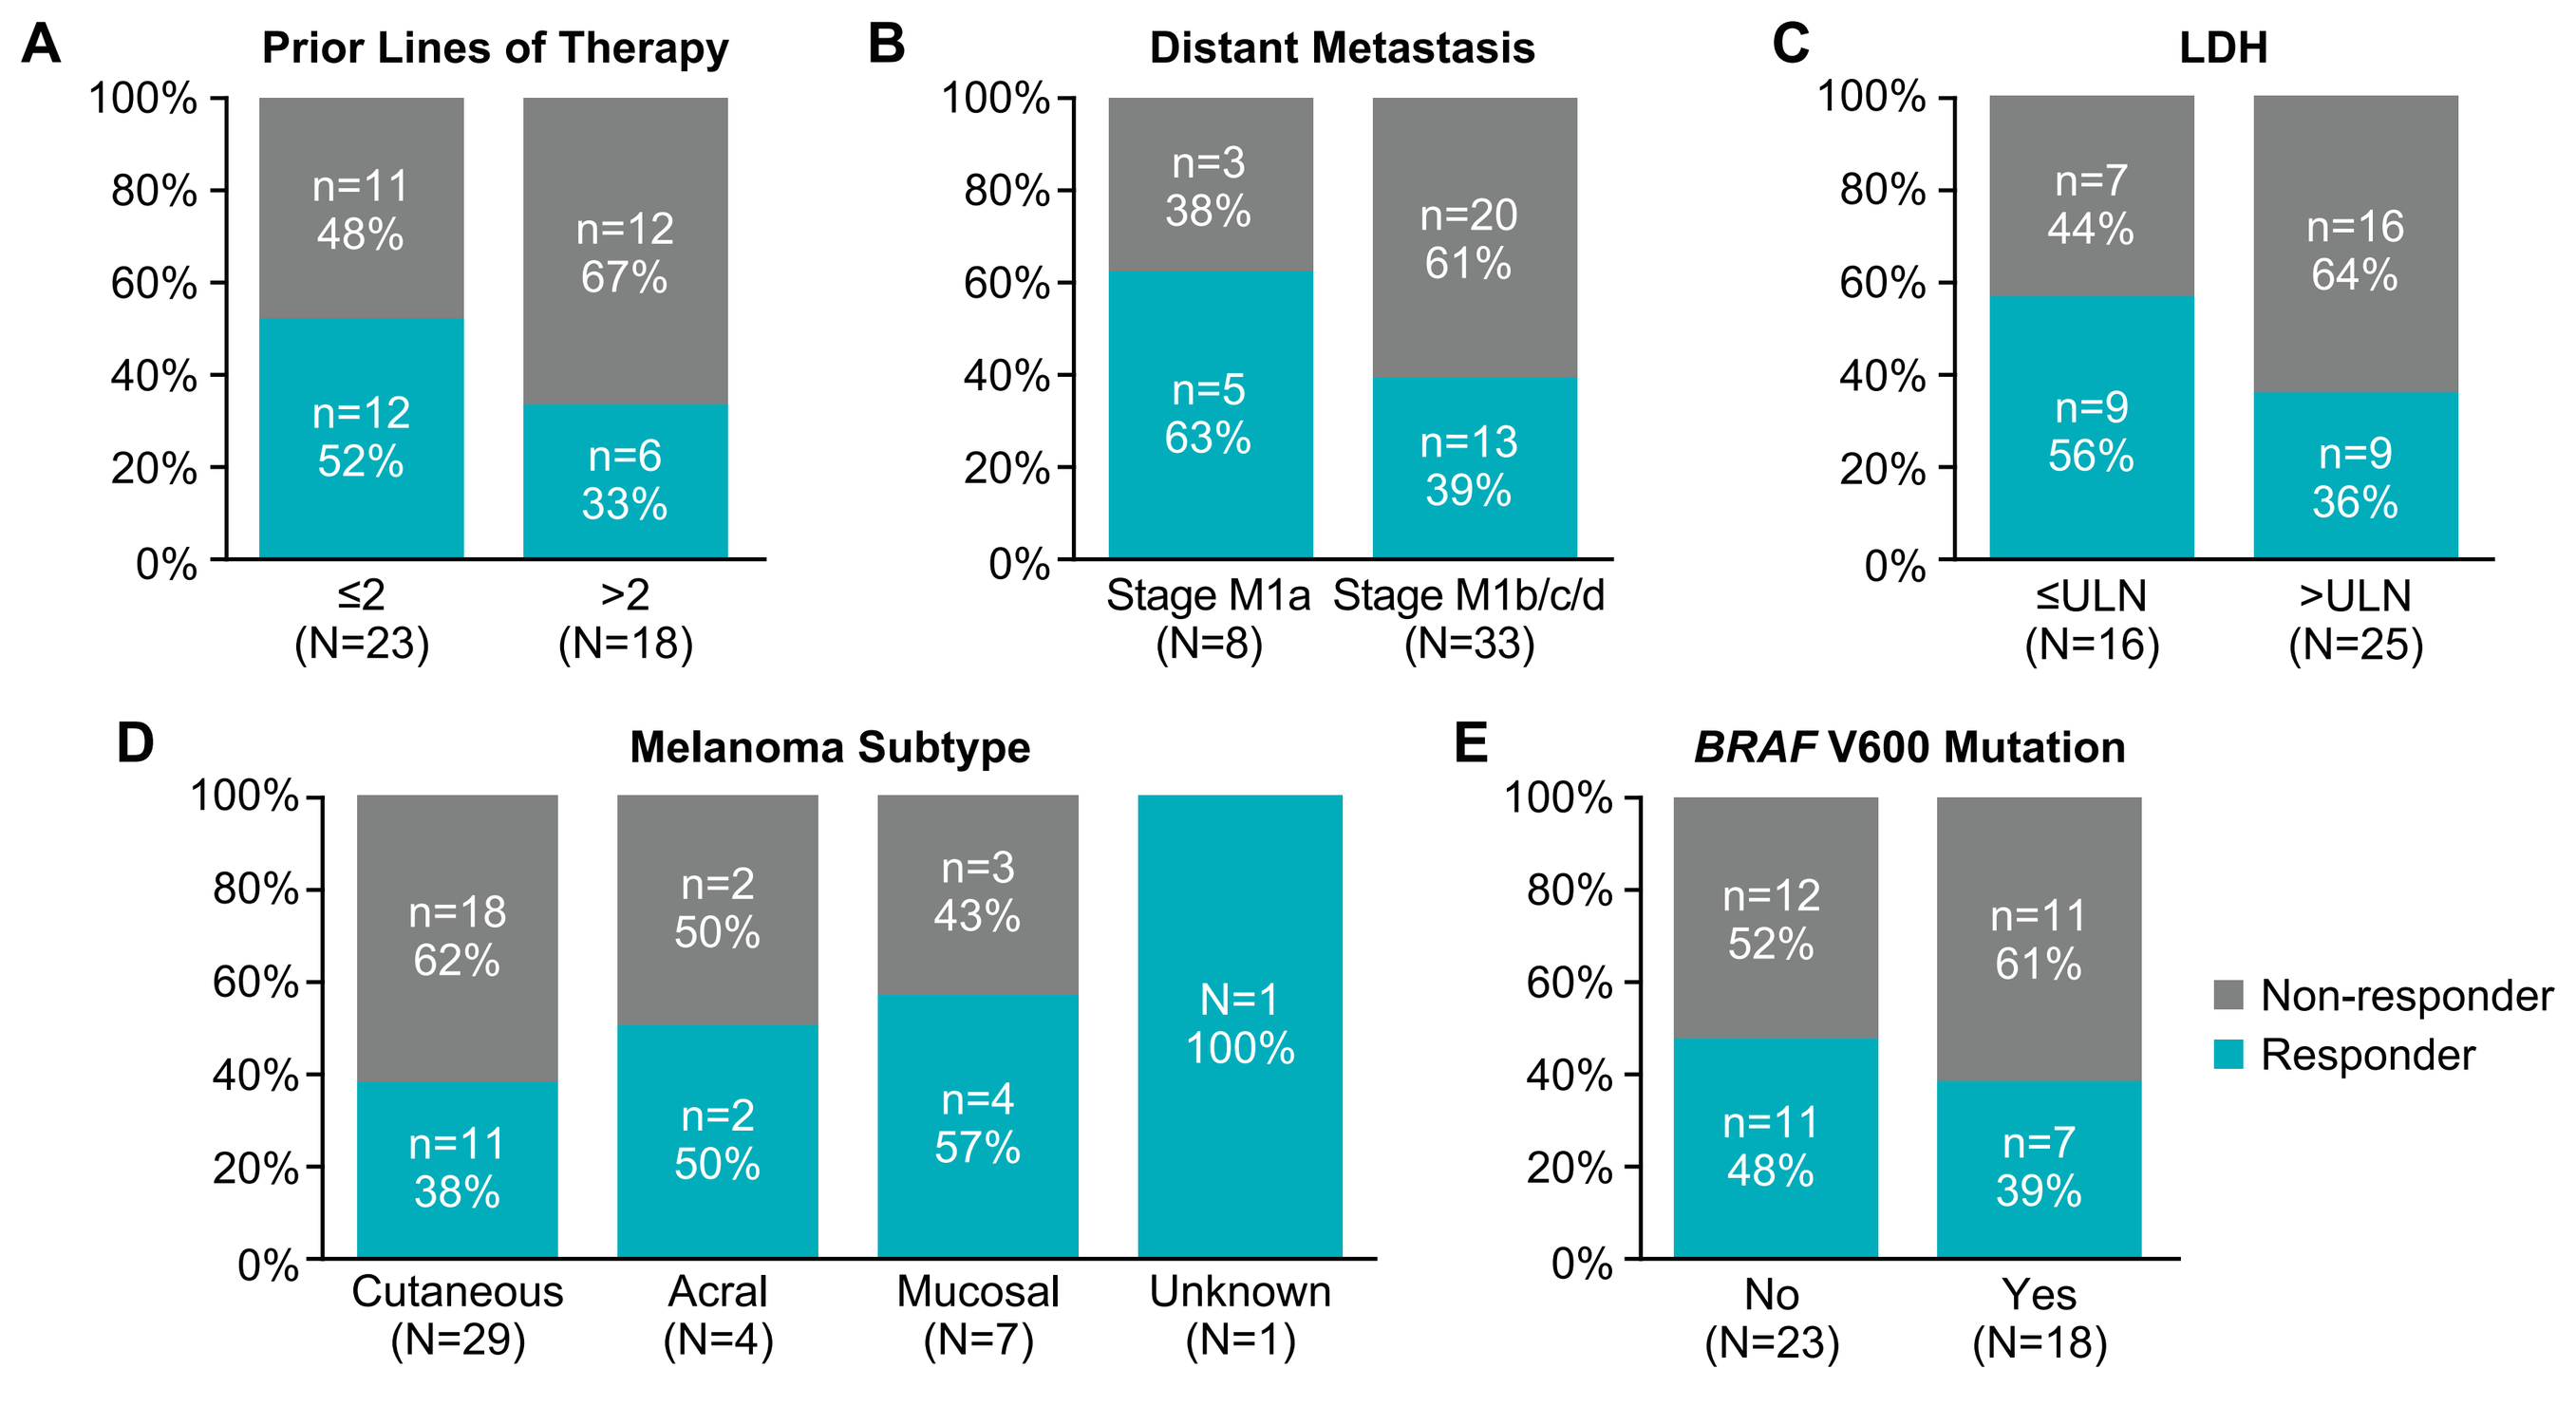

A Prior Lines of Therapy
≤2 (N=23): n=11 48%; n=12 52%
>2 (N=18): n=12 67%; n=6 33%
B Distant Metastasis
Stage M1a (N=8): n=3 38%; n=5 63%
Stage M1b/c/d (N=33): n=20 61%; n=13 39%
C LDH
≤ULN (N=16): n=7 44%; n=9 56%
>ULN (N=25): n=16 64%; n=9 36%
D Melanoma Subtype
Cutaneous (N=29): n=18 62%; n=11 38%
Acral (N=4): n=2 50%; n=2 50%
Mucosal (N=7): n=3 43%; n=4 57%
Unknown (N=1): N=1 100%
E BRAF V600 Mutation
No (N=23): n=12 52%; n=11 48%
Yes (N=18): n=11 61%; n=7 39%
Non-responder
Responder

(A) Response rates by number of prior lines of anti-cancer therapy. Patients were categorized as having received <2 or ≥2 prior systemic therapies before lifileucel. **(B)** Response rates by disease stage at enrollment. Bars depict proportions of responders and non-responders. **(C)** Response rates by LDH level at baseline. Patients were grouped as ≤ULN, and >ULN. Bars represent the distribution of responders and non-responders within each LDH category. **(D)** Response rates by melanoma subtype. Stacked bars show proportions of responders (CR/PR) and non-responders (SD/PD) across cutaneous non-acral, acral, mucosal, and unknown primary melanoma. **(E)** Response rates by *BRAF* V600 mutation status. Comparison of outcomes in patients with *BRAF*-mutant versus *BRAF* wild-type disease.

- Subgroup analysis revealed fewer prior lines of therapy, lower disease burden, and LDH ≤ULN associated with a greater responses to lifileucel (**Figure 3**)
 - Patients with ≤2 prior lines of therapy had an ORR of 52% (12/23), and those with >2 had an ORR of 33% (6/18)
 - Patients with M1a disease or LDH ≤ULN demonstrated higher response rates (63% and 56%, respectively) to TIL therapy compared to those with more advanced disease (39%) and elevated LDH (36%)
 - Patients across all melanoma subtypes achieved therapeutic response, with a 57% (4/7) ORR in patients with mucosal melanoma
 - Patients with *BRAF* V600 mutations had an ORR of 39% (7/18), and those with wild-type tumors had an ORR of 48% (11/23)

Figure 4. Tumor Harvest Site, TIL Characteristics, and IL-2 Dosing in Relation to Clinical Response

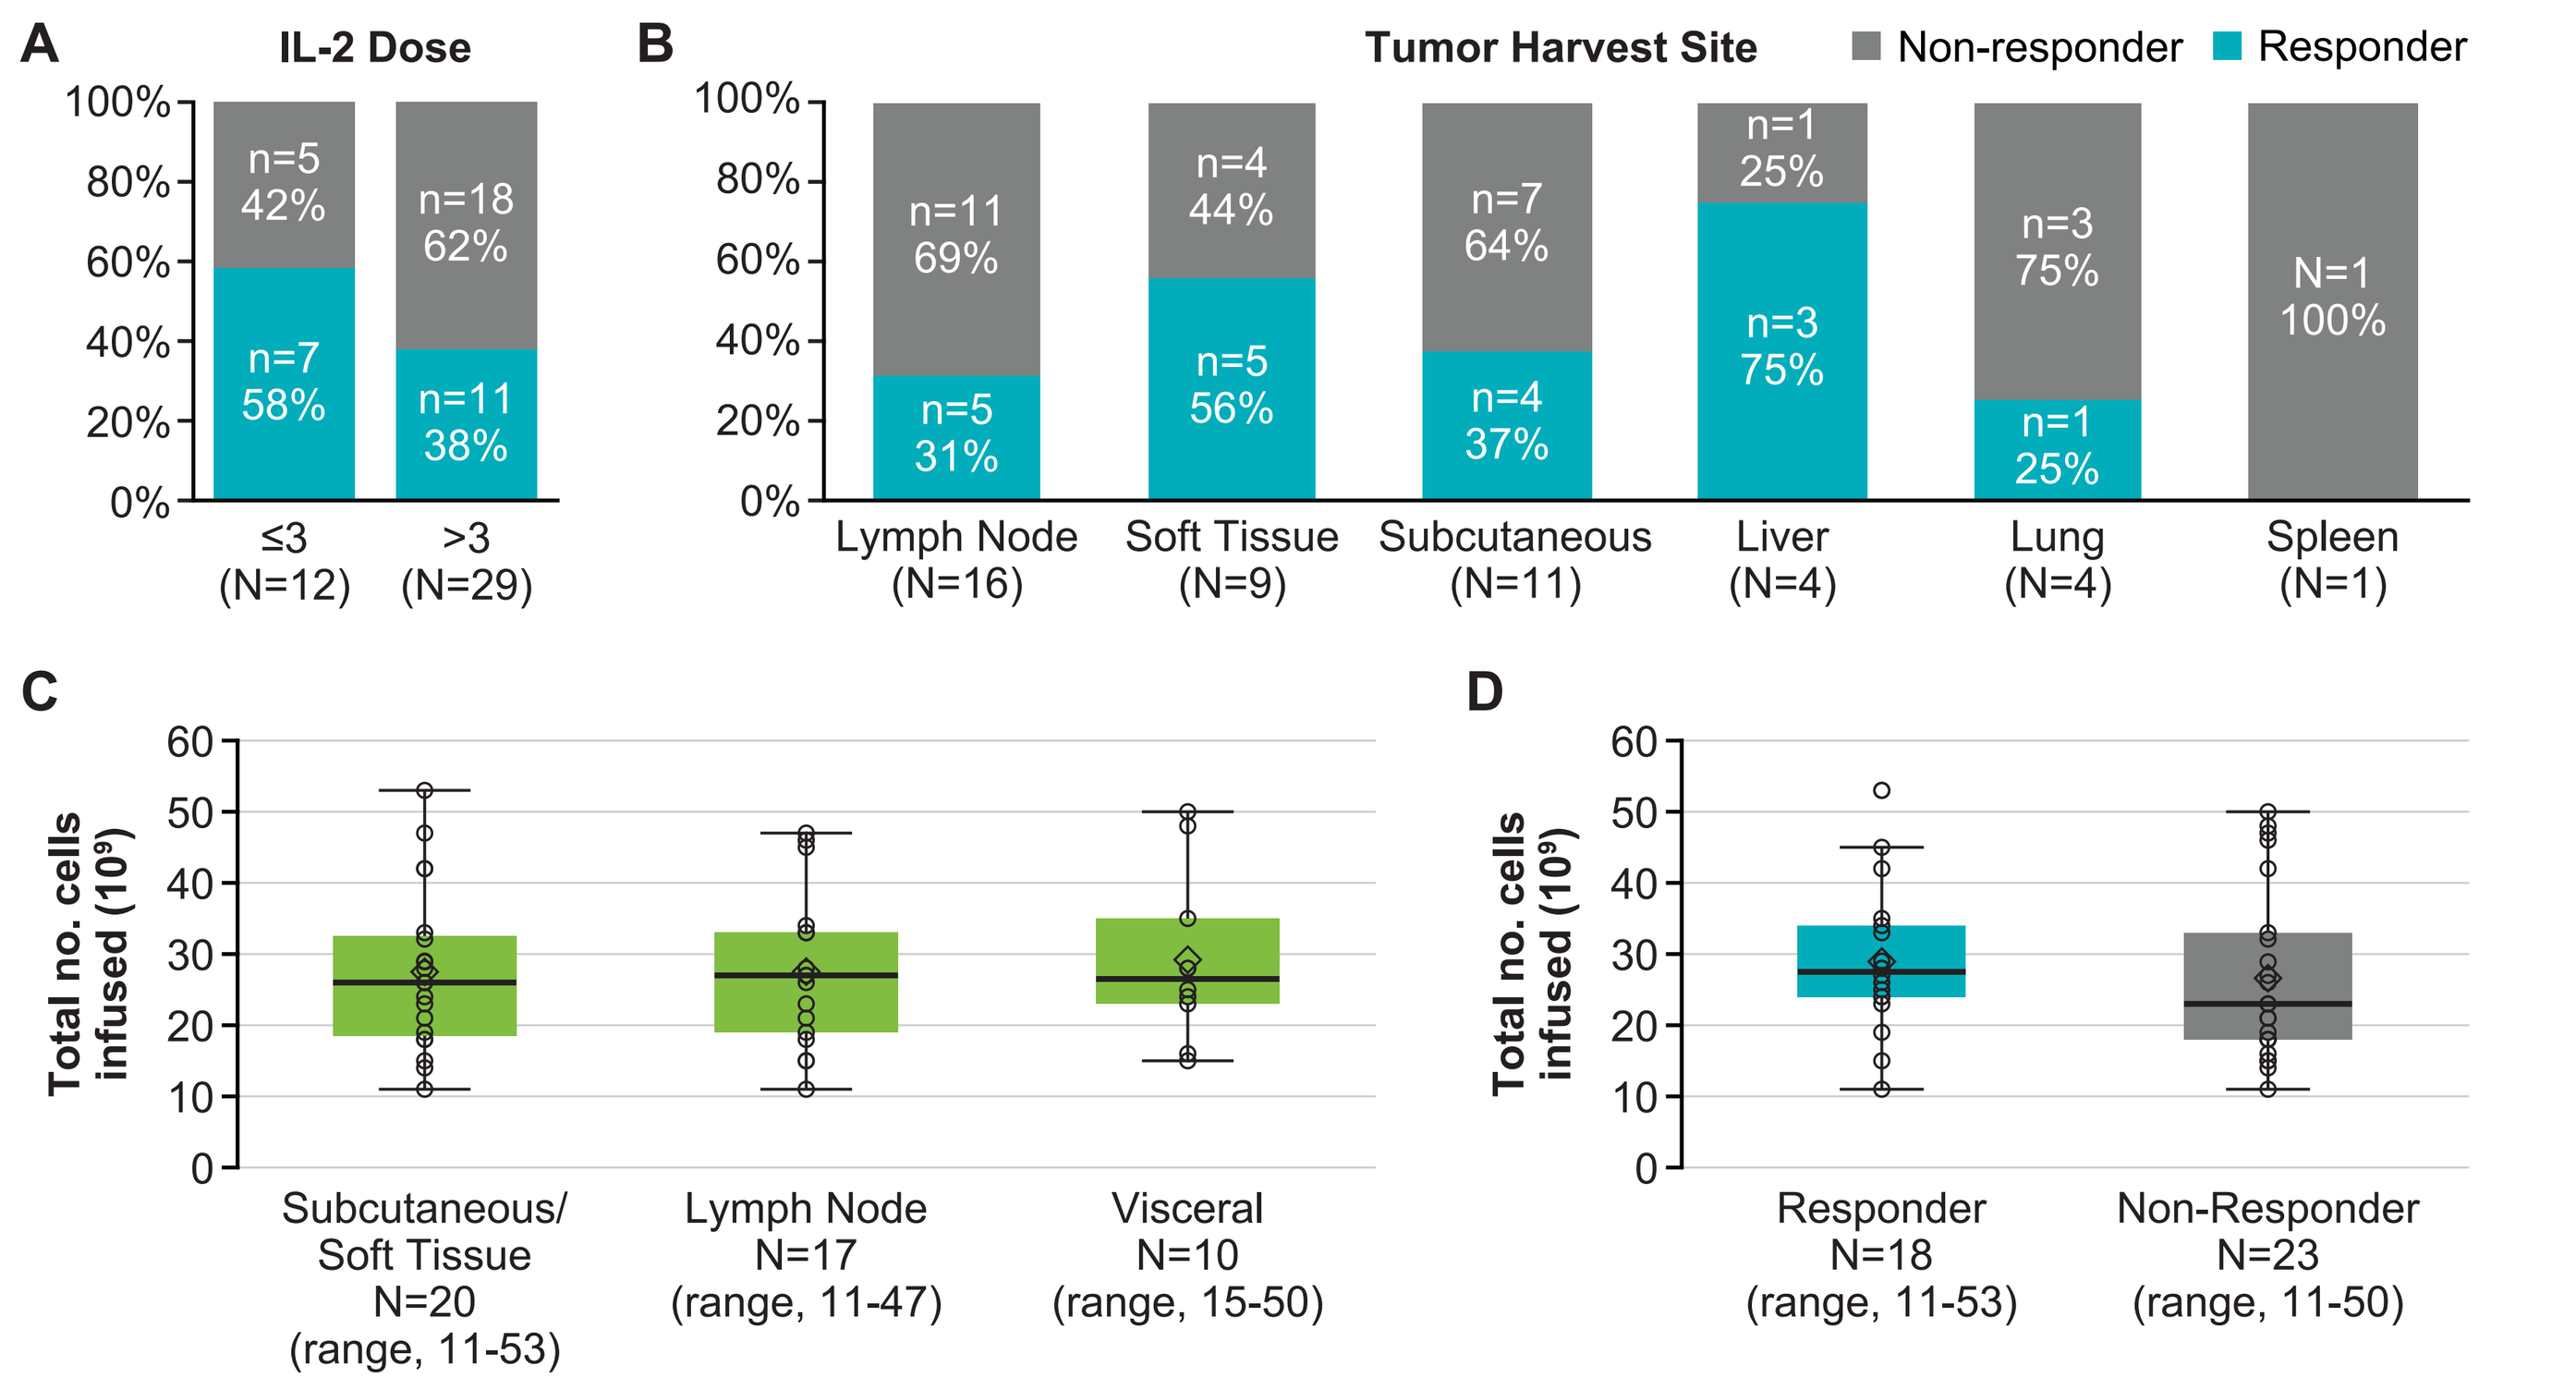

A IL-2 Dose
≤3 (N=12): n=5 42%; n=7 58%
>3 (N=29): n=18 62%; n=11 38%
B Tumor Harvest Site
Non-responder
Responder
Lymph Node (N=16): n=11 69%; n=5 31%
Soft Tissue (N=9): n=4 44%; n=5 56%
Subcutaneous (N=11): n=7 64%; n=4 37%
Liver (N=4): n=1 25%; n=3 75%
Lung (N=4): n=3 75%; n=1 25%
Spleen (N=1): N=1 100%
C
Total no. cells infused (10⁹)
Subcutaneous/ Soft Tissue N=20 (range, 11-53)
Lymph Node N=17 (range, 11-47)
Visceral N=10 (range, 15-50)
D
Total no. cells infused (10⁹)
Responder N=18 (range, 11-53)
Non-Responder N=23 (range, 11-50)

(A) Clinical response according to number of IL-2 doses administered. Stacked bar chart illustrates responder and non-responder proportions across IL-2 dosing groups. **(B)** Distribution of clinical response by tumor harvest site. Stacked bar chart demonstrates the proportions of responders and non-responders across anatomic sites used for tumor harvest. **(C)** Total viable of infused cells by tumor harvest site. Box-and-whisker plot depicts infused TIL counts (×10^9) in those harvested from subcutaneous or soft tissue (n=20), lymph node (n=17), and visceral sites (n=10). **(D)** Total viable number of infused TILs by response category. Box-and-whisker plot depicts infused TIL counts (×10^9) in responders (n=18) and non-responders (n=23).

- The ORR was 58% among patients who received ≤3 doses of IL-2 compared with 38% among those who received >3 doses of IL-2 (**Figure 4A**)
- Responses were comparable across tumor harvest sites (**Figure 4B**)
- Total infused cell yields were broadly similar across lesions, and there was no significant difference in the total number of infused TILs between responders and non-responders (**Figure 4C, D**)

Figure 5. Progression-Free Survival and Overall Survival Following TIL Therapy

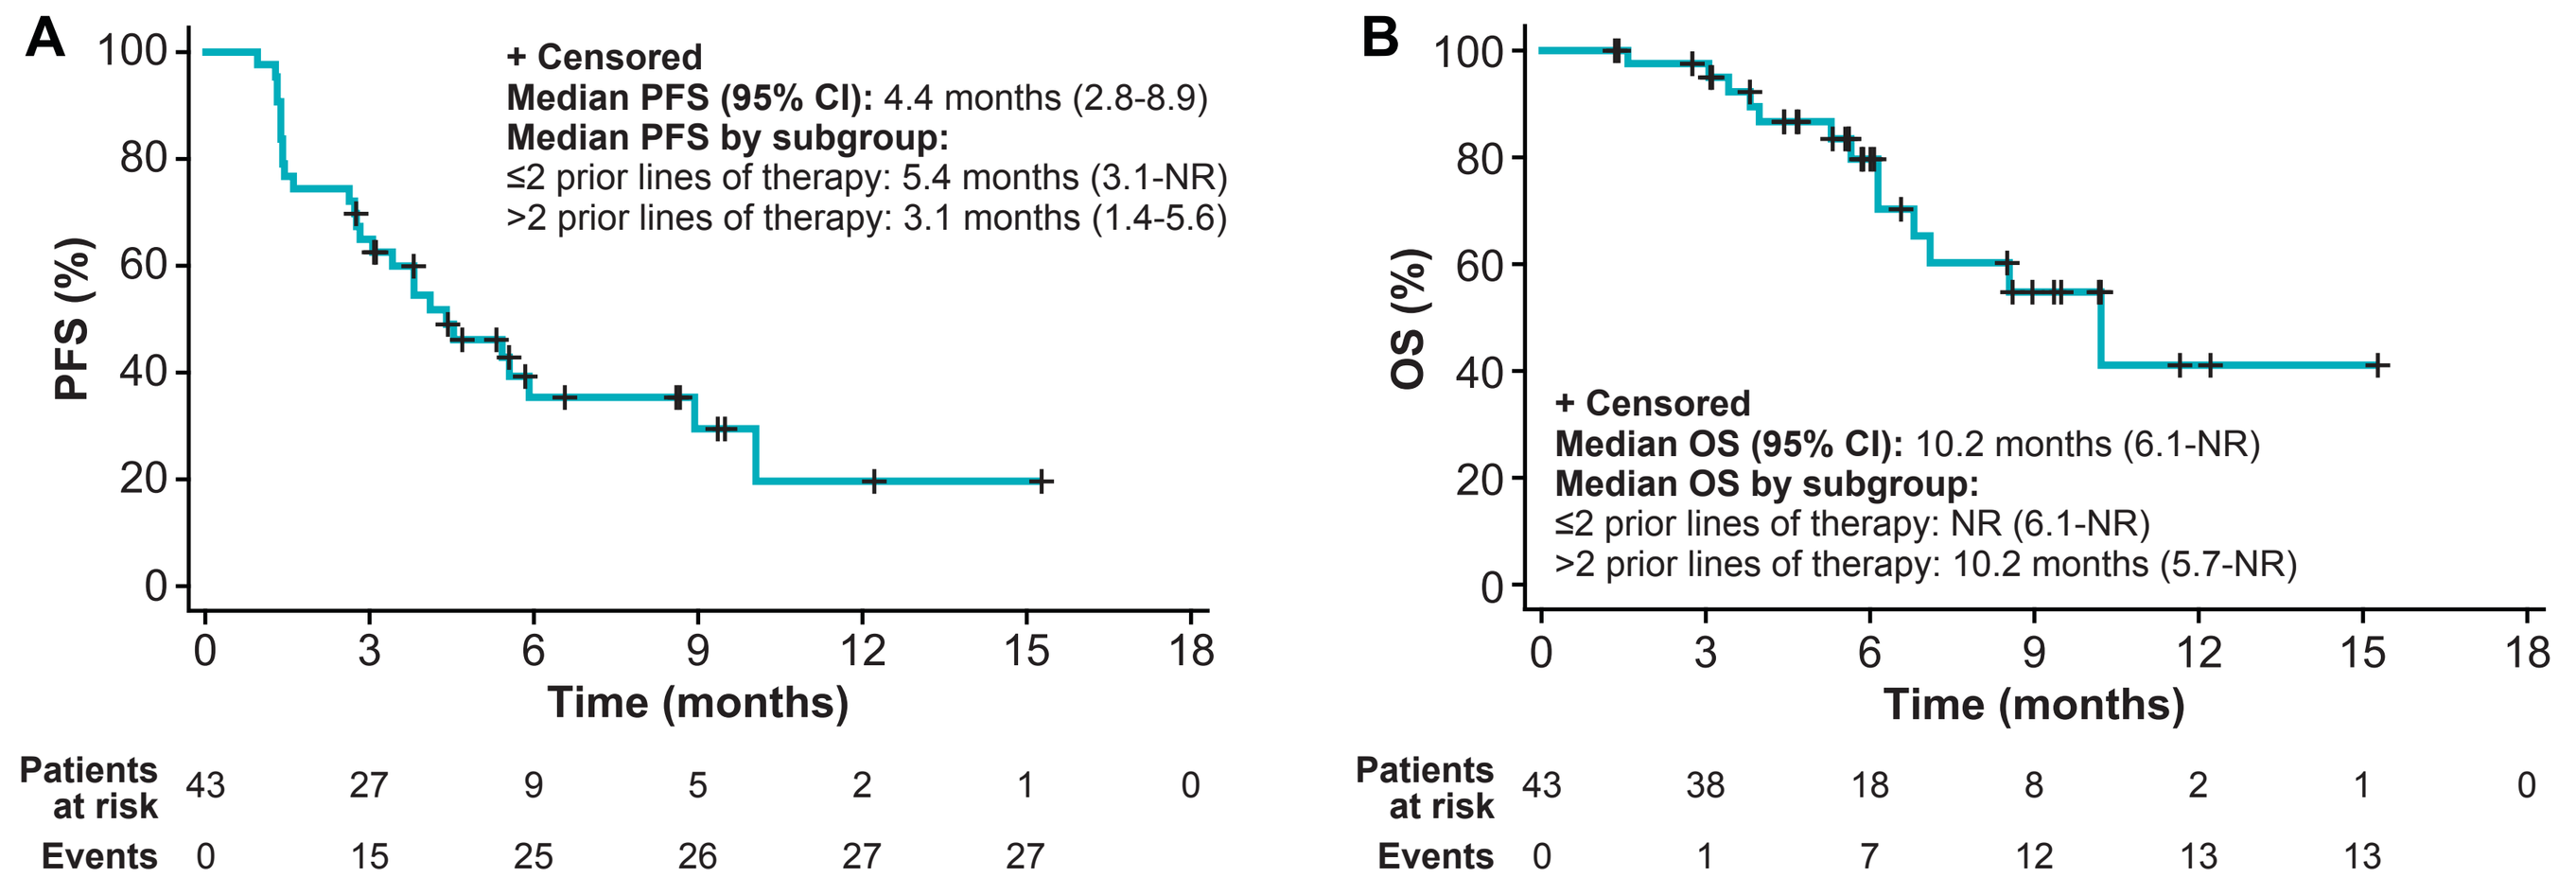

A
PFS (%)
+ Censored
Median PFS (95% CI): 4.4 months (2.8-8.9)
Median PFS by subgroup:
≤2 prior lines of therapy: 5.4 months (3.1-NR)
>2 prior lines of therapy: 3.1 months (1.4-5.6)
Time (months)
Patients at risk 43 27 9 5 2 1 0
Events 0 15 25 26 27 27
B
OS (%)
+ Censored
Median OS (95% CI): 10.2 months (6.1-NR)
Median OS by subgroup:
≤2 prior lines of therapy: NR (6.1-NR)
>2 prior lines of therapy: 10.2 months (5.7-NR)
Time (months)
Patients at risk 43 38 18 8 2 1 0
Events 0 1 7 12 13 13

(A) PFS from the time of TIL infusion. KM curve demonstrating PFS for the overall cohort (N=43). The median PFS was 4.4 months (95% CI, 2.8-8.9). The number of participants at risk at each time point and events are shown below the curve. **(B)** OS from the time of TIL infusion. KM curve depicting OS for the overall cohort (N=43). The median OS was 10.2 months (95% CI, 6.1-NR). The number of patients at risk and events at each time point are displayed below the curve.

- Medians for PFS and OS were 4.4 months (95% CI: 2.8-8.9) and 10.2 months (95% CI: 6.1-NR), respectively (**Figure 5**)

Figure 6. Progression-Free Survival by Subgroup

Subgroup	N	Event	HR (95% CI)	*P* Value
BRAF V600 mutation				
Yes	19	14	Ref	
No	24	13	0.508 (0.237-1.085)	0.08
Disease stage				
M1a	9	4	Ref	
M1b/c/d	34	23	1.268 (0.433-3.714)	0.66
Prior lines of therapy (metastatic)				
≤2	25	13	Ref	
>2	18	14	1.725 (0.809-3.675)	0.16
Total viable cell count				
<Median	20	12	Ref	
≥Median	23	15	1.184 (0.553-2.539)	0.66
Dose of IL-2				
≤3	12	8	Ref	
>3	31	19	0.780 (0.332-1.828)	0.57
Melanoma subtype				
Cutaneous	30	19	Ref	
Acral	4	3	0.939 (0.276-3.199)	0.92
Mucosal	8	4	0.539 (0.181-1.599)	0.26
Unknown	1	1	0.537 (0.067-4.309)	0.56
LDH				
≤ULN	17	7	Ref	
>ULN	26	20	2.386 (1.004-5.667)	0.05

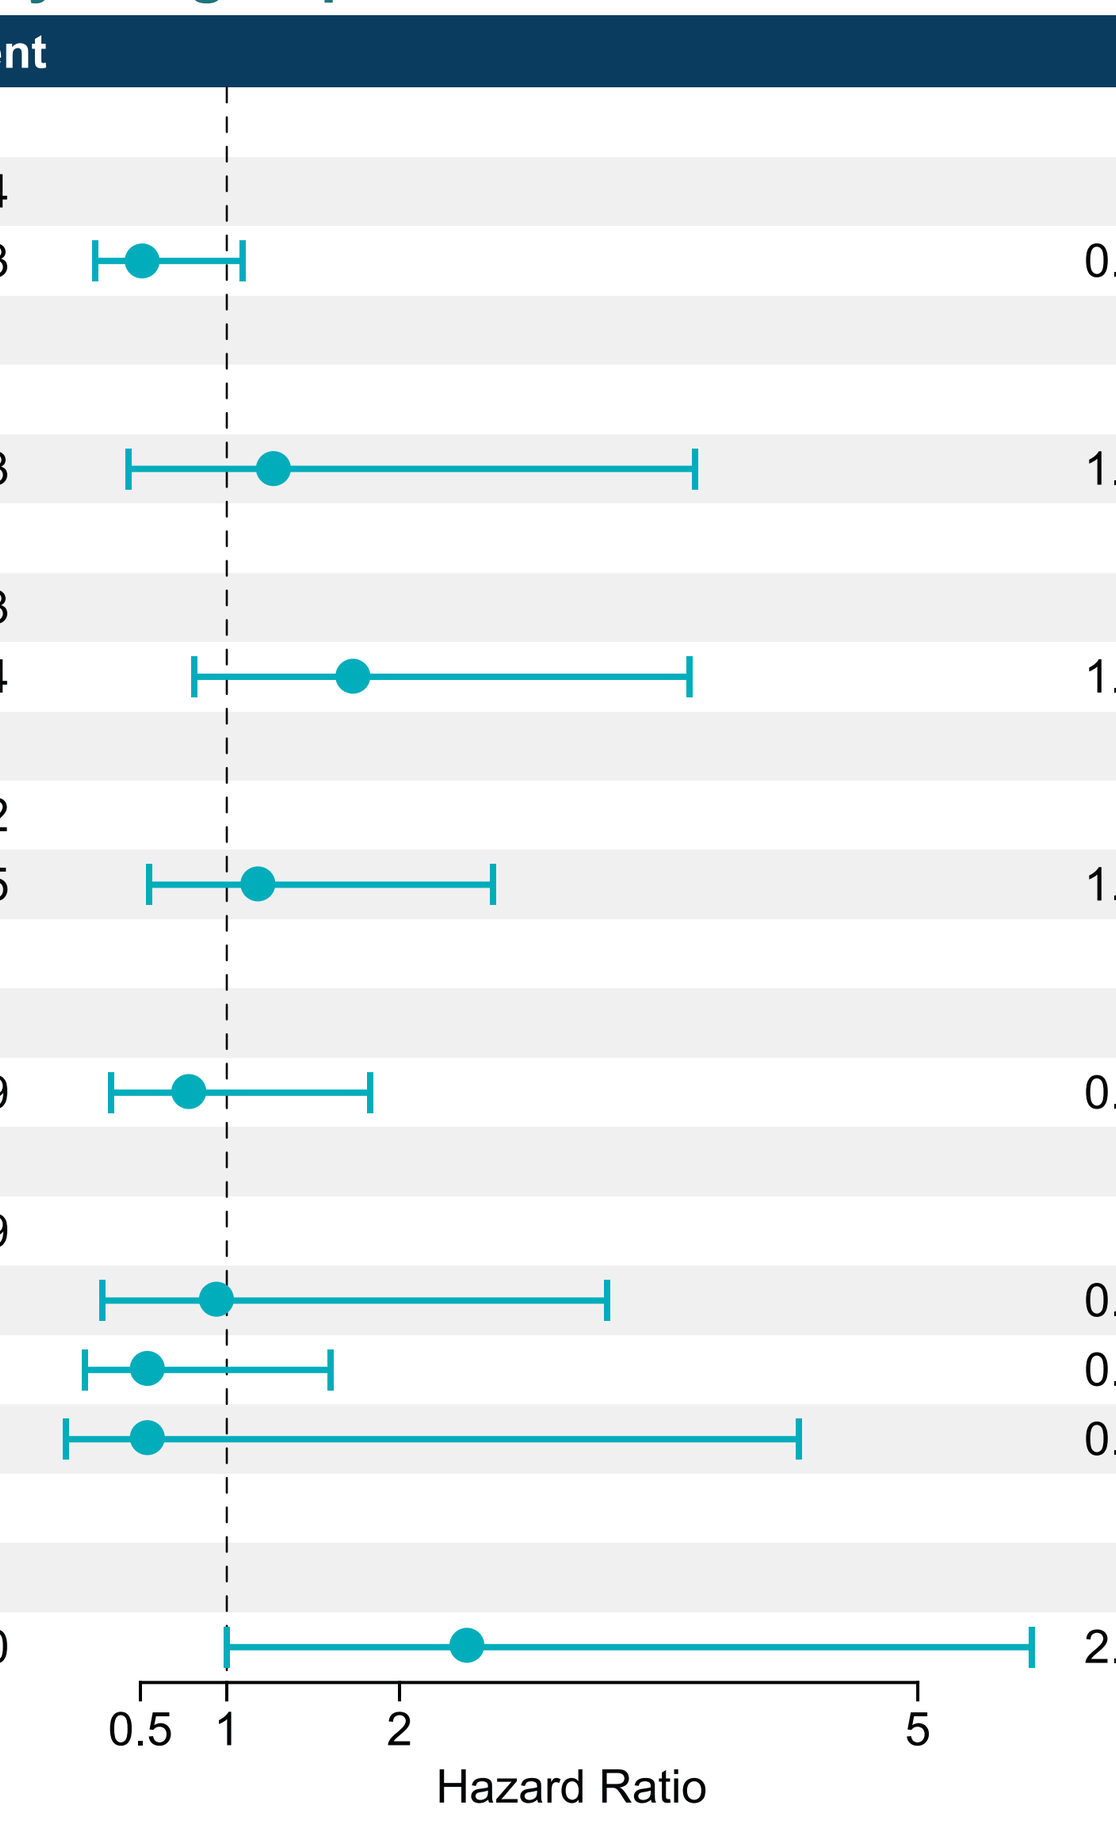

0.5
1
2
5
Hazard Ratio

- PFS was broadly similar across clinically relevant subgroups (**Figure 6**)
 - Elevated LDH (>ULN) was associated with significantly worse PFS compared to LDH ≤ULN (*P*=0.05)

Figure 7. Reasons for IL-2 Discontinuation

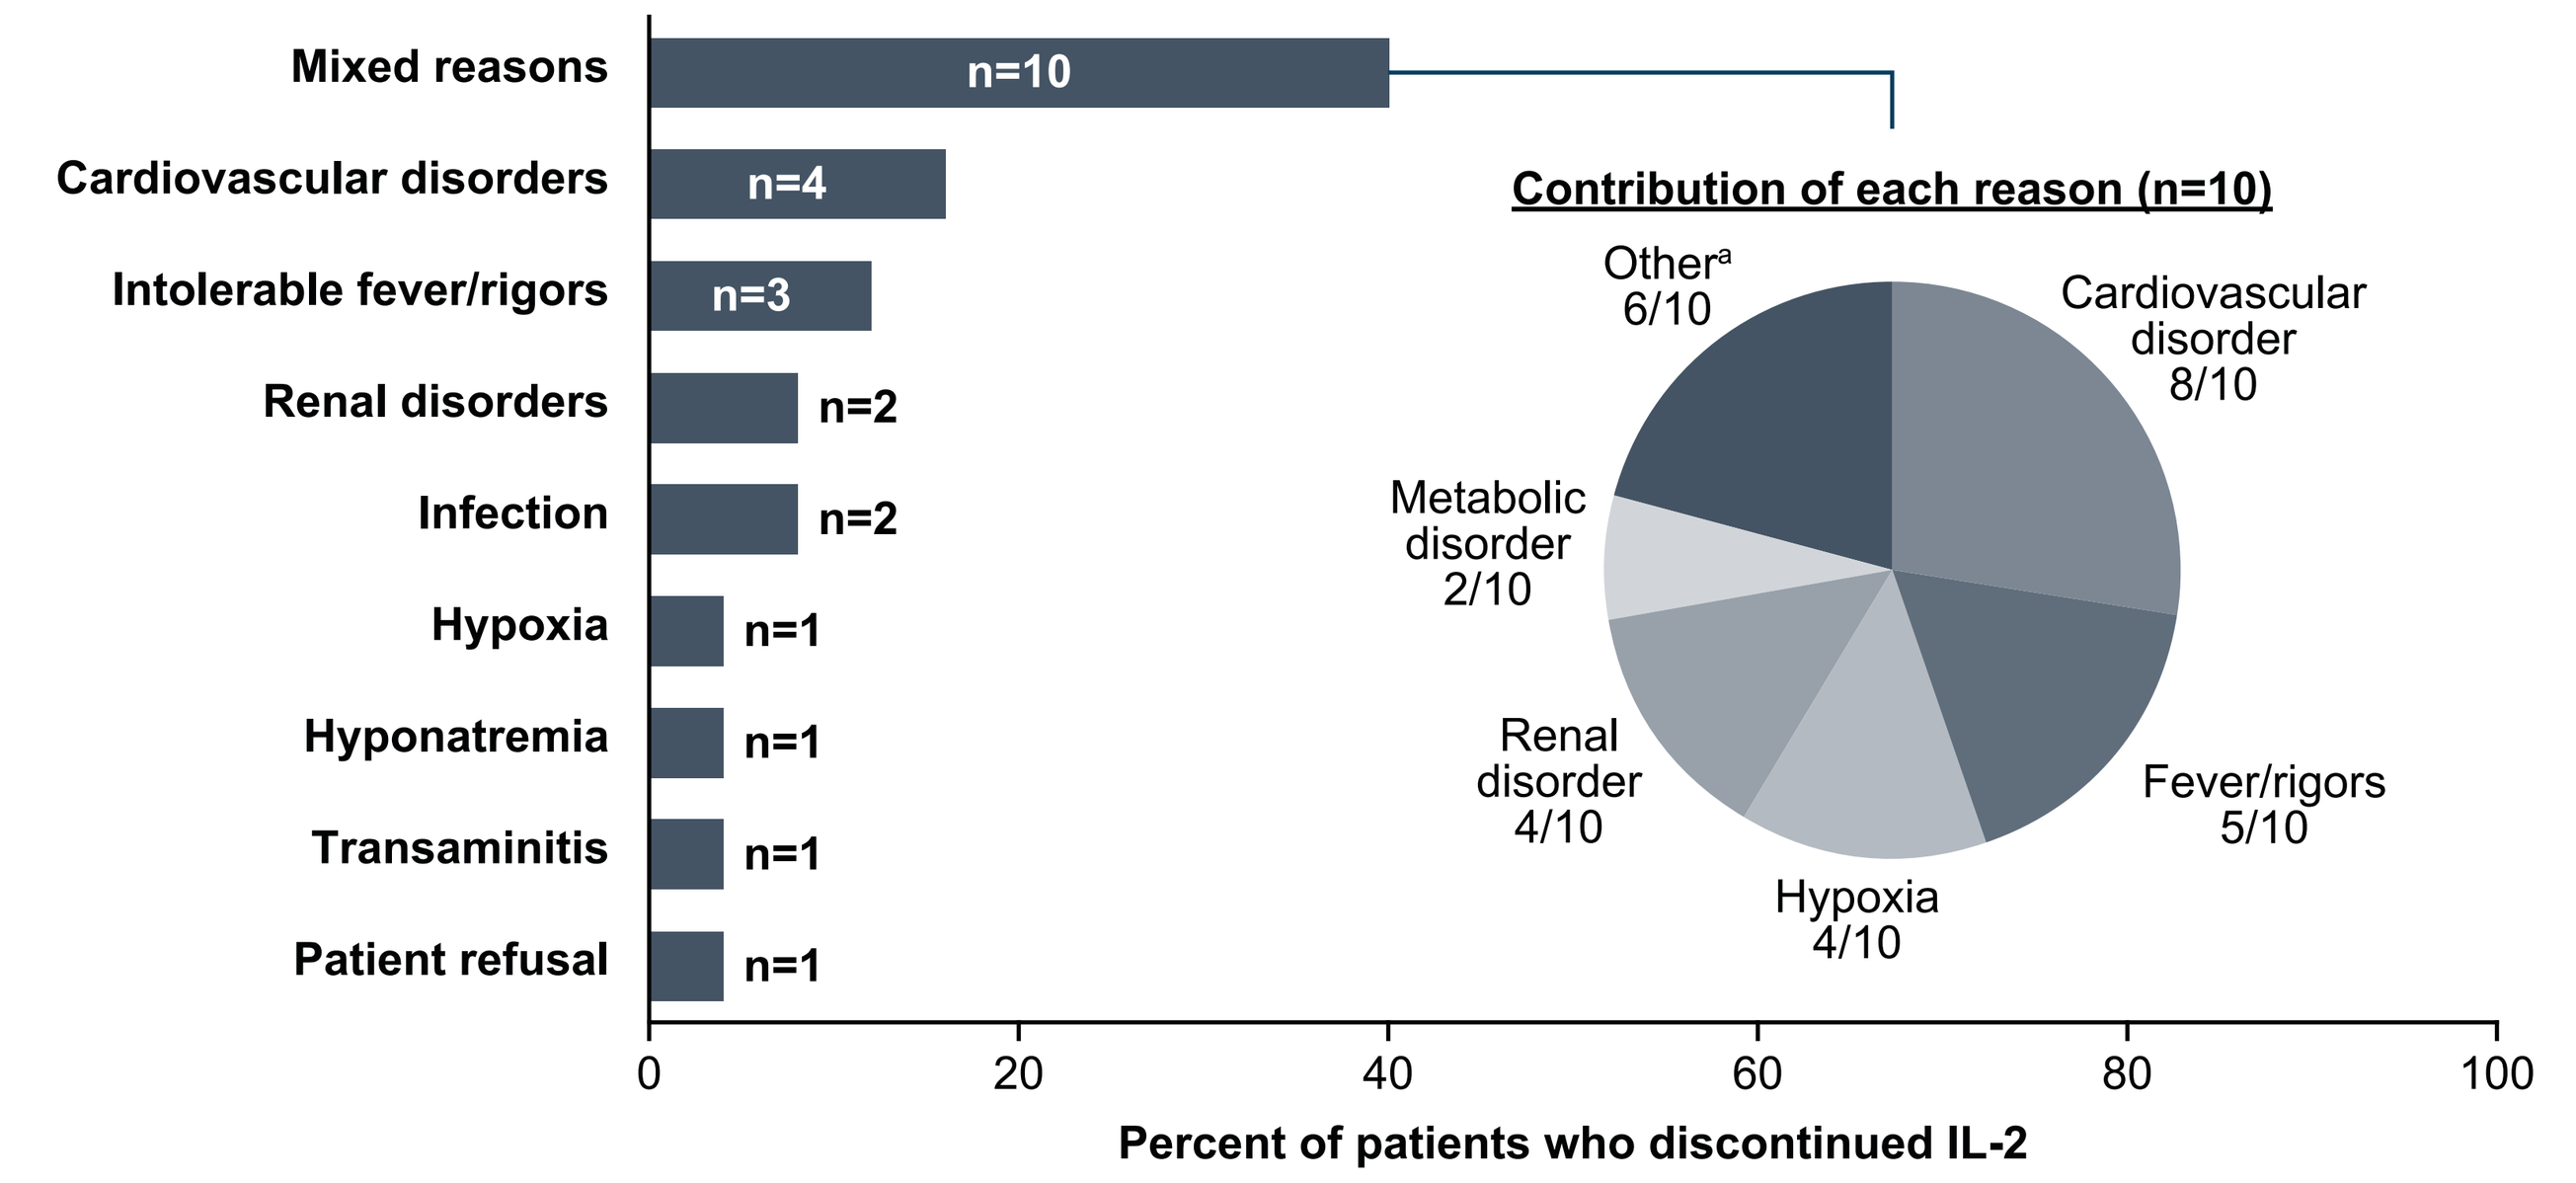

Mixed reasons n=10
Cardiovascular disorders n=4
Intolerable fever/rigors n=3
Renal disorders n=2
Infection n=2
Hypoxia n=1
Hyponatremia n=1
Transaminitis n=1
Patient refusal n=1
Percent of patients who discontinued IL-2
Contribution of each reason (n=10)
Cardiovascular disorder 8/10
Fever/rigors 5/10
Hypoxia 4/10
Renal disorder 4/10
Metabolic disorder 2/10
Other[a] 6/10

[a] Includes 1 each of altered mental state, gastrointestinal bleeding, COVID infection, volume overload, seizure-like activity, and edema.

- Patients were hospitalized for a median of 13 days (range, 4–50)
- IL-2 discontinuation commonly occurred due to fevers/rigors, cardiovascular AEs, hypoxia, and/or volume overload (**Figure 7**)
- One patient did not complete all planned doses of Cy due to AEs (hypotension and fatigue), and 3 did not complete all planned doses of Flu due to emergent surgical decompression (n=1), tachycardia (n=1), and *Clostridium difficile* colitis (n=1)
- No deaths were recorded within 45 days of TIL infusion

References

1. Sarnaik AA, et al. *J Clin Oncol*. 2021;39(24):2656-2666.
2. Chesney J, et al. *J Immunother Cancer*. 2022;10(12):e005755.
3. Medina T, et al. *J Clin Oncol*. 2025;43(33):3565-3572.
4. Amtagvi. Prescribing information. Iovance Biotherapeutics, Inc; 2024.
5. Karapetyan L, et al. *Hematol Oncol Clin North Am*. 2021;35(1):1-27.
6. Knight A, et al. *Cancers (Basel)*. 2023;15(4):1106.
7. Tawbi HA, et al. *N Engl J Med*. 2022;386(1):24-34.
8. Wolchok JD, et al. *N Engl J Med*. 2025;392(1):11-22.
9. Olson DJ, et al. *J Clin Oncol*. 2021;39(24):2647-2655.
10. Menzies AM, et al. *N Engl J Med*. 2022;386(17):1668-1669.
11. Ascierto PA, et al. *J Clin Oncol*. 2023;41(15):2724-2735.
12. Atkins MB, et al. *J Clin Oncol*. 2023;41(2):186-197.
13. Schadendorf D, et al. *Eur J Cancer*. 2024;204:114073.

Acknowledgments

- The authors thank the patients and their families who are participating in the study
- This study is sponsored by:
 - Moffitt Cancer Center - Cutaneous Oncology Department Immune Cell Therapy Program, Nikhil I. Khushalani, Amod A. Sarnaik, John E. Mullinax, Ricardo J. Gonzalez, Jonathan S. Zager, Matthew C. Perez, Vernon K Sondak, Shari Pilon-Thomas
 - Memorial Sloan Kettering Cancer Center - MSKCC Cellular Therapy Service, Charlotte Ariyan, Ned Bartlett, Carrie Bellerive, Danielle Bello, Erika Castaneda, Jeffrey Kuhn, Alyssa Sebba
 - Weill Cornell Medical Center - NYP Weill Cornell BMT Cell Therapy Program, Christopher Hackett, Gaurav Varma, Jessica Palmer
 - Honor Health Research Institute and Arizona State University John Shufedlt School of Medicine and Medical Engineering
 - Iovance Biotherapeutics, Inc. (San Carlos, CA, USA)
- Editorial support was provided by Second City Science and funded by Iovance. Editorial support and critical review were provided by Kavitha Kuppusamy, PhD

Abbreviations

AE, adverse event; *BRAF*, B-Raf proto-oncogene, serine/threonine kinase; CR, complete response; Cy, cyclophosphamide; DCR, disease control rate; Flu, fludarabine; HR, hazard ratio; ICI, immune checkpoint inhibitor; IL-2, interleukin-2; IV, intravenous; KM, Kaplan-Meier; LDH, lactate dehydrogenase; MEK, mitogen-activated protein kinase; NR, not reached; ORR, objective response rate; OS, overall survival; PD, progressive disease; PFS, progression-free survival; PR, partial response; q8-12hr, every 8 to 12 hours; Ref, reference; SD, stable disease; TIL, tumor-infiltrating lymphocyte; ULN, upper limit of normal.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the authors.