1107

IOV-END-201: a phase 2, multicenter, open-label study of lifileucel (tumor-infiltrating lymphocytes) in participants with previously treated advanced endometrial cancer

Amir Jazaeri,[1] Koji Matsuo,[2] Emese Zsiros,[3] Jason Chesney,[4] John Nakayama,[5] Akiko Suzuki,[6] Anjali Desai,[6] Siyu Zhu,[6] Brian Gastman,[6] Jeffrey Chou,[6] Friedrich Graf Finckenstein,[6] Kathleen Moore[7]

[1]University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]Keck School of Medicine, University of Southern California, Los Angeles, CA, USA; [3]Roswell Park Comprehensive Cancer Center, Buffalo, NY, USA; [4]UofL Health Brown Cancer Center, University of Louisville, Louisville, KY, USA; [5]Allegheny Health Network, Pittsburgh, PA, USA; [6]Iovance Biotherapeutics Inc, San Carlos, CA, USA; [7]Stephenson Cancer Center at the University of Oklahoma HSC, Oklahoma City, OK, USA

ANNUAL MEETING ON WOMEN'S CANCER
SEATTLE, WA • 2025

Background

- Treatment options are limited for patients with advanced endometrial cancer (EC) that has progressed during or after frontline platinum-based chemotherapy and anti-programmed cell death protein-1/programmed death ligand-1 (anti–PD-[L]1) therapy
- Anti–PD-(L)1 agents are standard-of-care therapy for patients with EC with both deficient mismatch repair (dMMR) and proficient mismatch repair (pMMR) tumors[1]
- Lifileucel, a tumor-infiltrating lymphocyte (TIL) therapy, has demonstrated clinical responses across anti–PD-(L)1 responsive solid tumor types such as melanoma, non-small cell lung, cervical, and head and neck cancers, including in patients who have progressed on or after prior therapy with anti–PD-(L)1 agents[2-5]
- The positive correlation demonstrated between the presence of TIL in endometrial tumor specimens and patient outcomes supports the potential utility of an immunotherapeutic approach, such as autologous TIL, as treatment for EC[6-8]
- Successful ex vivo expansion of TIL from endometrial tumors was accomplished, and the TIL generated from endometrial tumors demonstrated antitumor activity in vitro.[9,10] These data support clinical investigation of TIL cell therapy in patients with EC
- Based on this, we hypothesized that lifileucel also may have antitumor activity in patients with advanced EC

Objective

- The IOV-END-201 study will enroll patients with dMMR or pMMR tumors to investigate the efficacy and safety of the lifileucel regimen in participants with previously treated endometrial cancer and explore the role of mismatch repair (MMR) status in response to treatment with TIL therapy

Patient Journey Overview

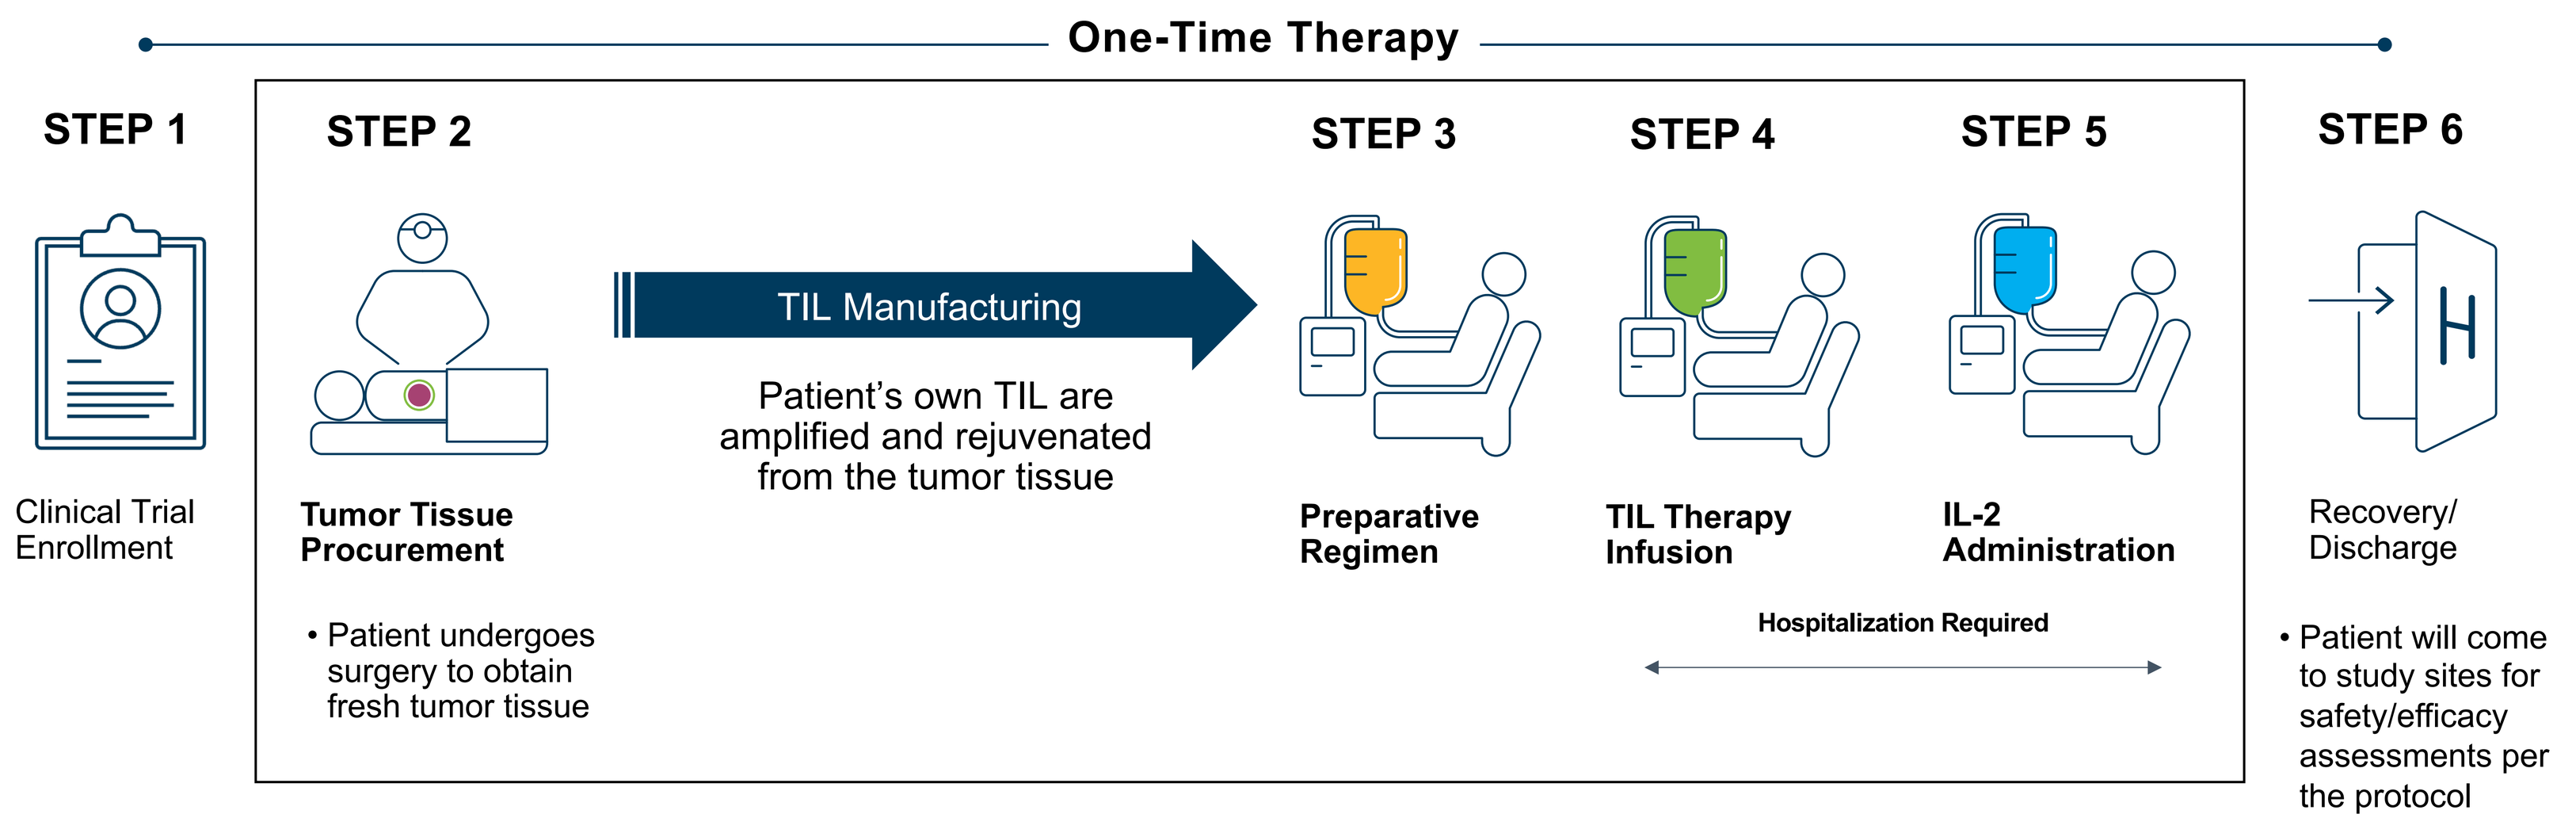
One-Time Therapy
STEP 1
Clinical Trial Enrollment
STEP 2
Tumor Tissue Procurement
• Patient undergoes surgery to obtain fresh tumor tissue
TIL Manufacturing
Patient's own TIL are amplified and rejuvenated from the tumor tissue
STEP 3
Preparative Regimen
STEP 4
TIL Therapy Infusion
STEP 5
IL-2 Administration
Hospitalization Required
STEP 6
Recovery/ Discharge
• Patient will come to study sites for safety/efficacy assessments per the protocol

IOV-END-201 Study Overview

- IOV-END-201 is a phase 2, open-label, single-arm, multicenter study (NCT06481592) evaluating the efficacy and safety of lifileucel and the role of MMR status in response to treatment with lifileucel in patients with advanced EC who previously received chemotherapy and an anti–PD-(L)1 agent sequentially or in combination (**Figure 1** and **Figure 2**)

Study Design and Treatment Regimen

Figure 1. IOV-END-201 Study Design

Patient Population

- Endometrial cancer previously treated with **platinum-based chemotherapy and anti–PD-(L)1 therapy** for recurrent, metastatic or primary unresectable disease
- **Up to 3 lines of prior systemic therapy with no more than 1 line of chemotherapy** for recurrent, metastatic or primary unresectable disease

Patients with pMMR tumors

Patients with dMMR tumors

Sample Size

- N=60 (n=30 per MMR subgroup)

Endpoints

- **Primary**: ORR per RECIST v1.1 as assessed by the investigator
- **Secondary**: complete response (CR) rate, duration of response (DOR), disease control rate (DCR), progression free survival (PFS), overall survival (OS), safety and tolerability
- **Exploratory**: Lifileucel manufacturing feasibility, *in vivo* T cell persistence, and correlative biomarkers

Figure 2. IOV-END-201 Treatment Schema

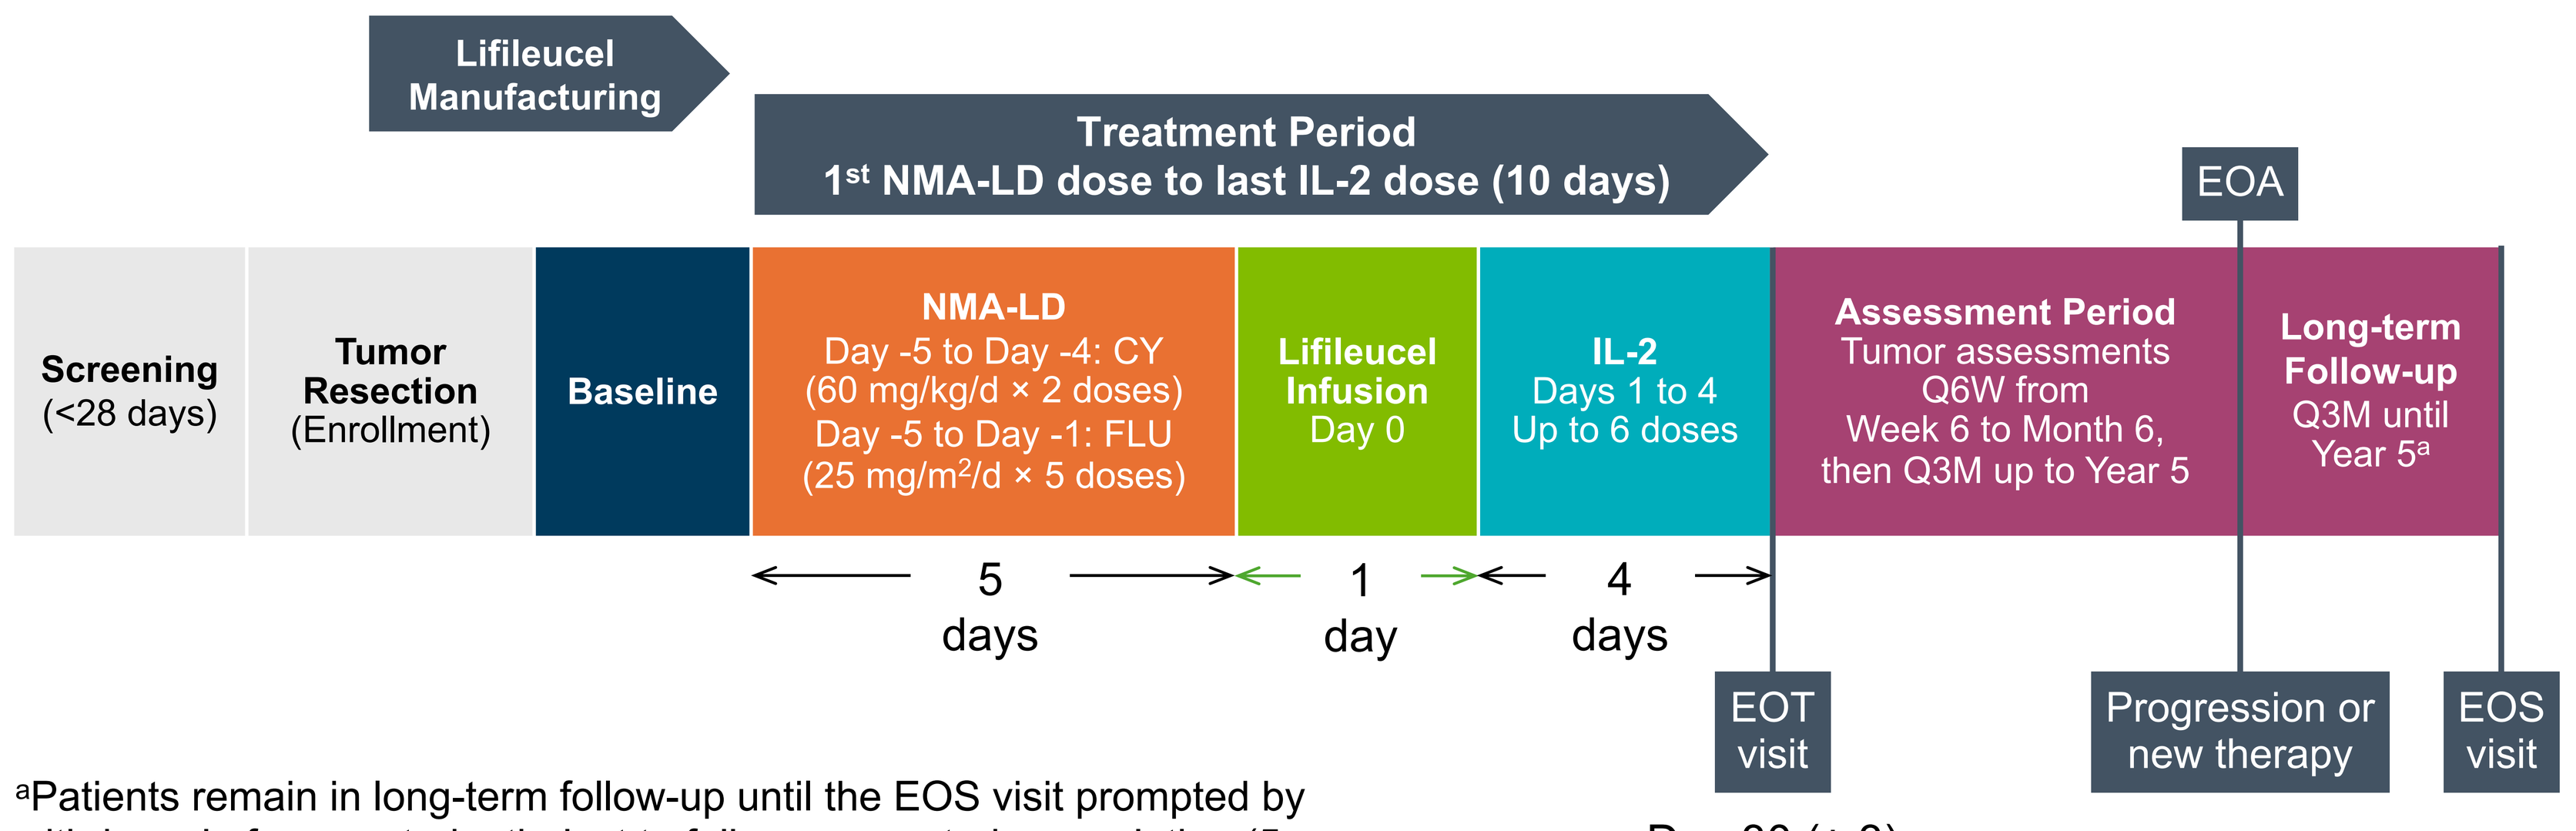
Lifileucel Manufacturing
Treatment Period
1st NMA-LD dose to last IL-2 dose (10 days)
EOA
Screening (<28 days)
Tumor Resection (Enrollment)
Baseline
NMA-LD Day -5 to Day -4: CY (60 mg/kg/d × 2 doses) Day -5 to Day -1: FLU (25 mg/m2/d × 5 doses)
Lifileucel Infusion Day 0
IL-2 Days 1 to 4 Up to 6 doses
Assessment Period Tumor assessments Q6W from Week 6 to Month 6, then Q3M up to Year 5
Long-term Follow-up Q3M until Year 5a
5 days
1 day
4 days
EOT visit
Progression or new therapy
EOS visit
Day 30 (± 3)

[a]Patients remain in long-term follow-up until the EOS visit prompted by withdrawal of consent, death, lost to follow-up, or study completion (5 years after lifileucel infusion).

Study Sites

- The study will be conducted across multiple sites in the United States (**Figure 3**)

Figure 3. IOV-END-201 Study Sites

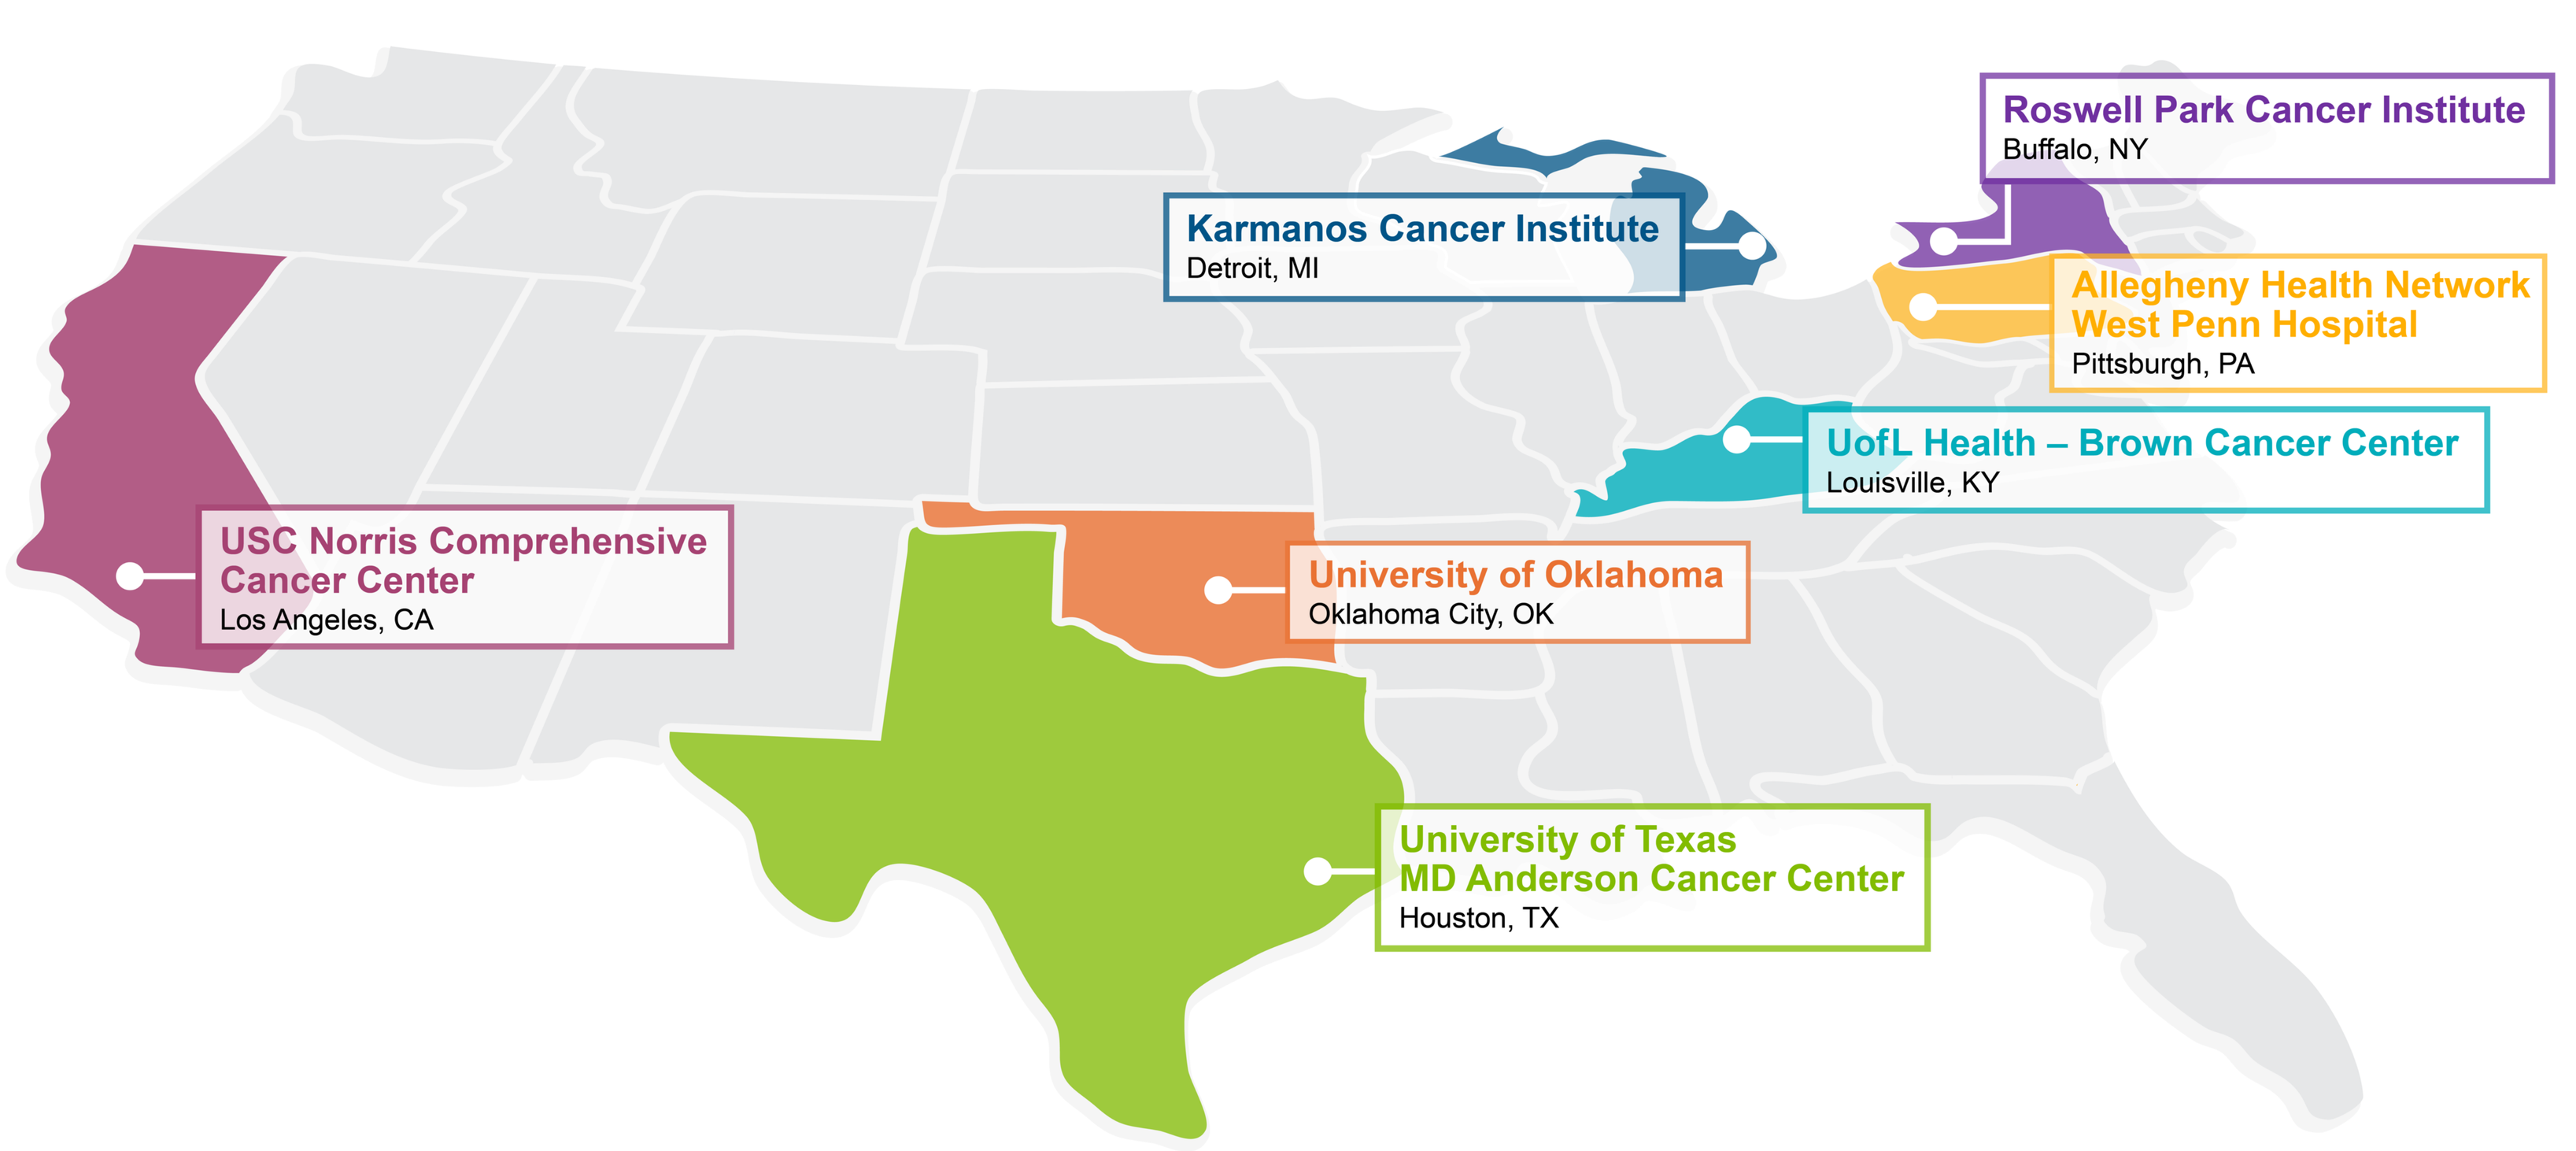
Roswell Park Cancer Institute
Buffalo, NY
Karmanos Cancer Institute
Detroit, MI
Allegheny Health Network West Penn Hospital
Pittsburgh, PA
UofL Health – Brown Cancer Center
Louisville, KY
USC Norris Comprehensive Cancer Center
Los Angeles, CA
University of Oklahoma
Oklahoma City, OK
University of Texas MD Anderson Cancer Center
Houston, TX

Key Eligibility Criteria

Inclusion Criteria

- Age ≥18 years; age >70 years permitted after discussion with the medical monitor
- Histologically confirmed advanced EC including carcinosarcoma (but not uterine sarcoma)
- Advanced EC that is not amenable to curative treatment with surgery and/or radiation therapy
- Prior therapy criteria:
 - Progression on or after platinum-based chemotherapy and anti–PD-(L)1 therapy or nonresponse or intolerance to those therapies
 - ≤3 lines of systemic therapy for recurrent, metastatic, or primary unresectable disease with ≤1 line of chemotherapy
 - Documented radiographic disease progression during or after the last line of therapy
- Eastern Cooperative Oncology Group (ECOG) performance status of 0–1 and an estimated life expectancy of >6 months
- ≥1 measurable lesion(s) in addition to resectable lesion(s) or aggregate lesions with an estimated minimum diameter of 1.5 cm for lifileucel generation
- Adequate organ function, including adequate cardiopulmonary function

Exclusion Criteria

- Symptomatic untreated brain metastases
- Active medical illnesses that would pose increased risks for study participation (eg, systemic infections; coagulation disorders; other active major medical illnesses of the cardiovascular, respiratory, or immune systems)
- Any form of primary or acquired immunodeficiency (eg, SCID or AIDS)
- Other primary malignancy in the last 3 years
- Prior organ allograft or cell therapy within the past 20 years
- Current systemic steroid therapy >10 mg/day of prednisone or another steroid equivalent dose

References

1. National Comprehensive Cancer Network. Uterine Neoplasms (Version 1.2025). **2.** Chesney J, et al. *J Immunother Cancer*. 2022;10:e005755. **3.** Creelan BC, et al. Presented at SITC 2024; November 6-10, 2024; Houston, TX. **4.** Jazaeri AA, et al. Presented at ASCO 2019; June 1-3, 2019; Chicago, IL. **5.** Jimeno A, et al. Presented at SITC 2020; November 11-14, 2020; Virtual. **6.** Kono-Sato T, et al. *BMC Cancer.* 2022;22:361. **7.** Liu Y, et al. *Oncotarget*. 2019;10(9):982–992. **8.** Fan CT, et al. *J Clin Med*. 2023;12:603. **9.** Fang J, et al. *Int J Gynecol Cancer*. 2024;34(Suppl 3):A173. **10.** Fang J, et al. Presented at IGCS 2024; October 16-18, 2024; Dublin, Ireland.

Abbreviations

AIDS, acquired immunodeficiency syndrome; CR, complete response; CY, cyclophosphamide; d, day; DCR, disease control rate; dMMR, deficient mismatch repair; DOR, duration of response; EC, endometrial cancer; ECOG, Eastern Cooperative Oncology Group; EOA, end of assessment; EOS, end of study; EOT, end of treatment; FLU, fludarabine; IL-2, interleukin-2; MMR, mismatch repair; NMA-LD, nonmyeloablative lymphodepletion; ORR, objective response rate; OS, overall survival; PD-1, programmed cell death protein-1; PD-L1, programmed cell death ligand 1; PFS, progression-free survival; pMMR, proficient mismatch repair; Q3M, every 3 months; Q6W, every 6 weeks; SCID, severe combined immunodeficiency disease; TIL, tumor-infiltrating lymphocytes.

Acknowledgments

This study was sponsored by Iovance Biotherapeutics, Inc (San Carlos, CA, USA).

We acknowledge Dr. Katherine Mager at Roswell Park Comprehensive Cancer Center and Dr. Robert Morris at Karmanos Cancer Institute for their support of new investigational sites.

Medical writing and editorial support was provided by Peloton Advantage, LLC, an OPEN Health company, and funded by Iovance.

Disclosures

Amir Jazaeri: AstraZeneca, AvengeBio, GlaxoSmithKline, Greenfire Bio, Immatics USA, Imunon, Iovance Biotherapeutics, Lilly, Lixtie Biotechnology, MacroGenics, Merck, Sentinel Bio, Stryker, Theolytics. **Koji Matsuo**: Nothing to disclose. **Emese Zsiros**: ArsenalBio, Merck, NextCure. **Jason Chesney**: Amgen, Iovance Biotherapeutics, Obsidian, Replimune. **John Nakayama**: AstraZeneca, Eisai, Iovance Biotherapeutics, Merck, Myrias. **Akiko Suzuki, Anjali Desai, Siyu Zhu, Brian Gastman, Jeffrey Chou**: Iovance Biotherapeutics. **Friedrich Graf Finckenstein**: Bristol Myers Squibb, Iovance Biotherapeutics, Roche. **Kathleen Moore**: AstraZeneca, Aadi, Aravive, Blueprint, Caris, Duality, Daiichi Sankyo, Eisai, GlaxoSmithKline, Genentech/Roche, ImmunoGen, I-Mab, Janssen, Lilly, Mereo, Merck, Mersana, Myriad, Novartis, OncXerna, Onconova, Panavance, Regeneron, Verastem, VBL Therapeutics, Zentalis.

Corresponding Author

Kathleen Moore: Kathleen-Moore@ouhsc.edu

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the authors

ClinicalTrials.gov
NCT06481592

Society of Gynecologic Oncology Annual Meeting on Women's Cancer, March 14–17, 2025, Seattle, WA, USA

MULTIPLY YOUR IMPACT