24 Presented at the Society for Immunotherapy of Cancer (SITC) Spring Scientific, Cellular Therapy for Solid Tumors | March 12–14, 2025 | San Diego, CA

Multimodal single-cell sequencing analysis reveals putative tumor-reactive population in lifileucel TIL products

Joe Dean[1], Theresa Medina[2], Amod Sarnaik[3], Jason Chesney[4], Mike Cusnir[5], Joe Yglesias[1], Behzad Damirchi[1], Mark Ozeck[1], Kranthi Kunkalla[1], Brian Gastman[1], Rana Fiaz[1], Giri Sulur[1], Hequn Yin[1], Rongsu Qi[1]

[1]Iovance Biotherapeutics Inc, San Carlos, California, USA; [2]University of Colorado Cancer Center–Anschutz Medical Campus, Aurora, CO, USA; [3]H Lee Moffitt Cancer Center and Research Institute, Tampa, FL, USA; [4]Brown Cancer Center, Louisville, KY, USA; [5]Mount Sinai Comprehensive Cancer Center, Miami Beach, FL, USA

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the authors.

For more information, please contact Joe Dean: joe.dean@iovance.com

Background

- Lifileucel is an FDA-approved autologous TIL cell therapy for the treatment of advanced melanoma[1]
- We previously presented assessment of TIL drug product (DP) at a single-cell (sc) level from 20 patients

Figure 1. Lifileucel Tumor-infiltrating Lymphocyte (TIL) Cell Therapy

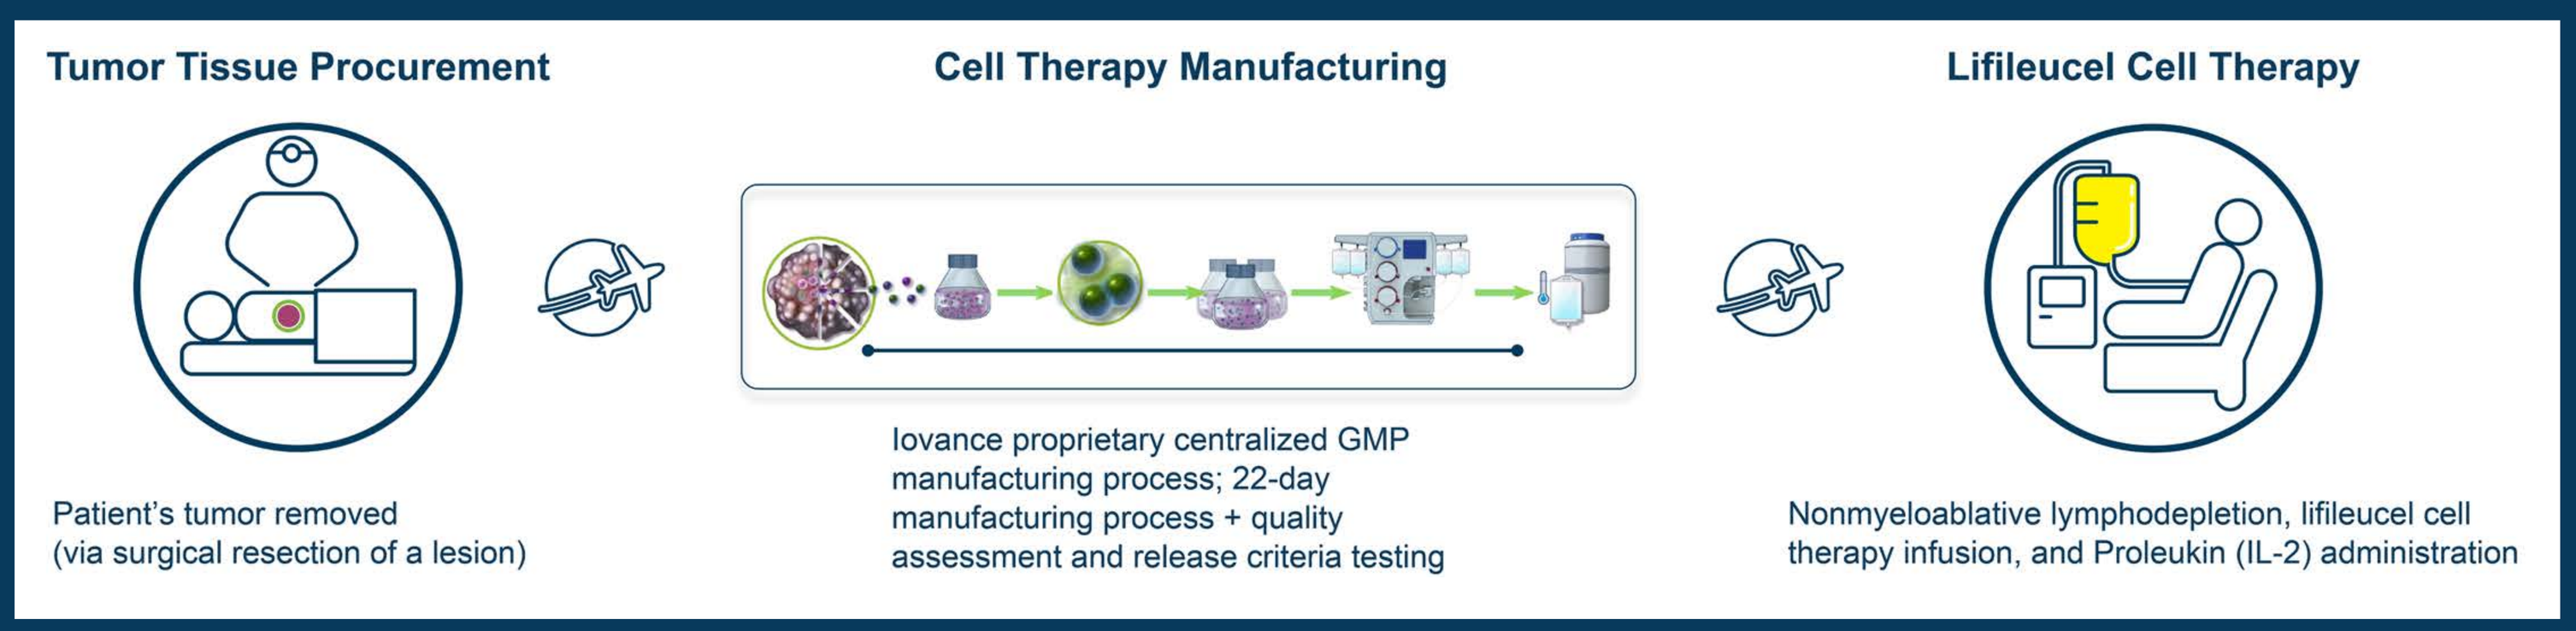
Tumor Tissue Procurement
Cell Therapy Manufacturing
Lifileucel Cell Therapy
Patient's tumor removed (via surgical resection of a lesion)
Iovance proprietary centralized GMP manufacturing process; 22-day manufacturing process + quality assessment and release criteria testing
Nonmyeloablative lymphodepletion, lifileucel cell therapy infusion, and Proleukin (IL-2) administration

- What are the potential cellular determinants of response to TIL cell therapy?

Objectives

- To further characterize lifileucel at a sc level using TIL DP from 34 patients and analyze baseline tumor digest T cells from a subset of these patients using high-dimension multimodal sequencing
- To characterize and compare putative tumor-reactive T cells in baseline tumor and TIL DP

Methods

- Cell barcode-matched scRNA-sequencing and scTCR-sequencing
 – 10x genomics 5' immune profiling
- C-144-01 metastatic melanoma dataset (NCT02360579)
 – Lifileucel, N=27
 – Matched tumor digest (CD45 enriched) and lifileucel, N=7
- Baseline tumor digest single-cell analysis
 – After data quality check and subsetting on T cells, sctransform[2,3] was used for normalization and feature selection, followed by PCA and Harmony[4] integration, clustering, and PaCMAP[5] dimensionality reduction visualization
 – Clusters were annotated manually based on marker genes, cell-type–specific gene signatures using VISION,[6] and automated annotation using scATOMIC[7]
 – Putative tumor-reactive T-cell clusters were identified based on expression level of the NeoTCR8 gene signature[8]
 – scTCR-Seq visualizations were generated using scRepertoire[9] and ggalluvial[10]
- Lifileucel single-cell analysis
 – To aid with cell-type annotation, CITE-Seq was performed on a subset of 4 patients (2 R, 1 SD, 1 PD)
 – Multimodal weighted-nearest neighbor analysis and manual cluster annotation were used to develop a single-cell TIL reference map
 – Gene set quantification was performed after sample level pseudo-bulking of NeoTCR8 clusters using GSVA[11]
- GraphPad Prism was used for summary data visualizations and statistics

Results

Figure 2. Profiles of Tumor T-Cell Subsets

- Putative tumor-reactive T-cell proportion in tumor appears to be associated with response (**Figure 2** and **Figure 3**)

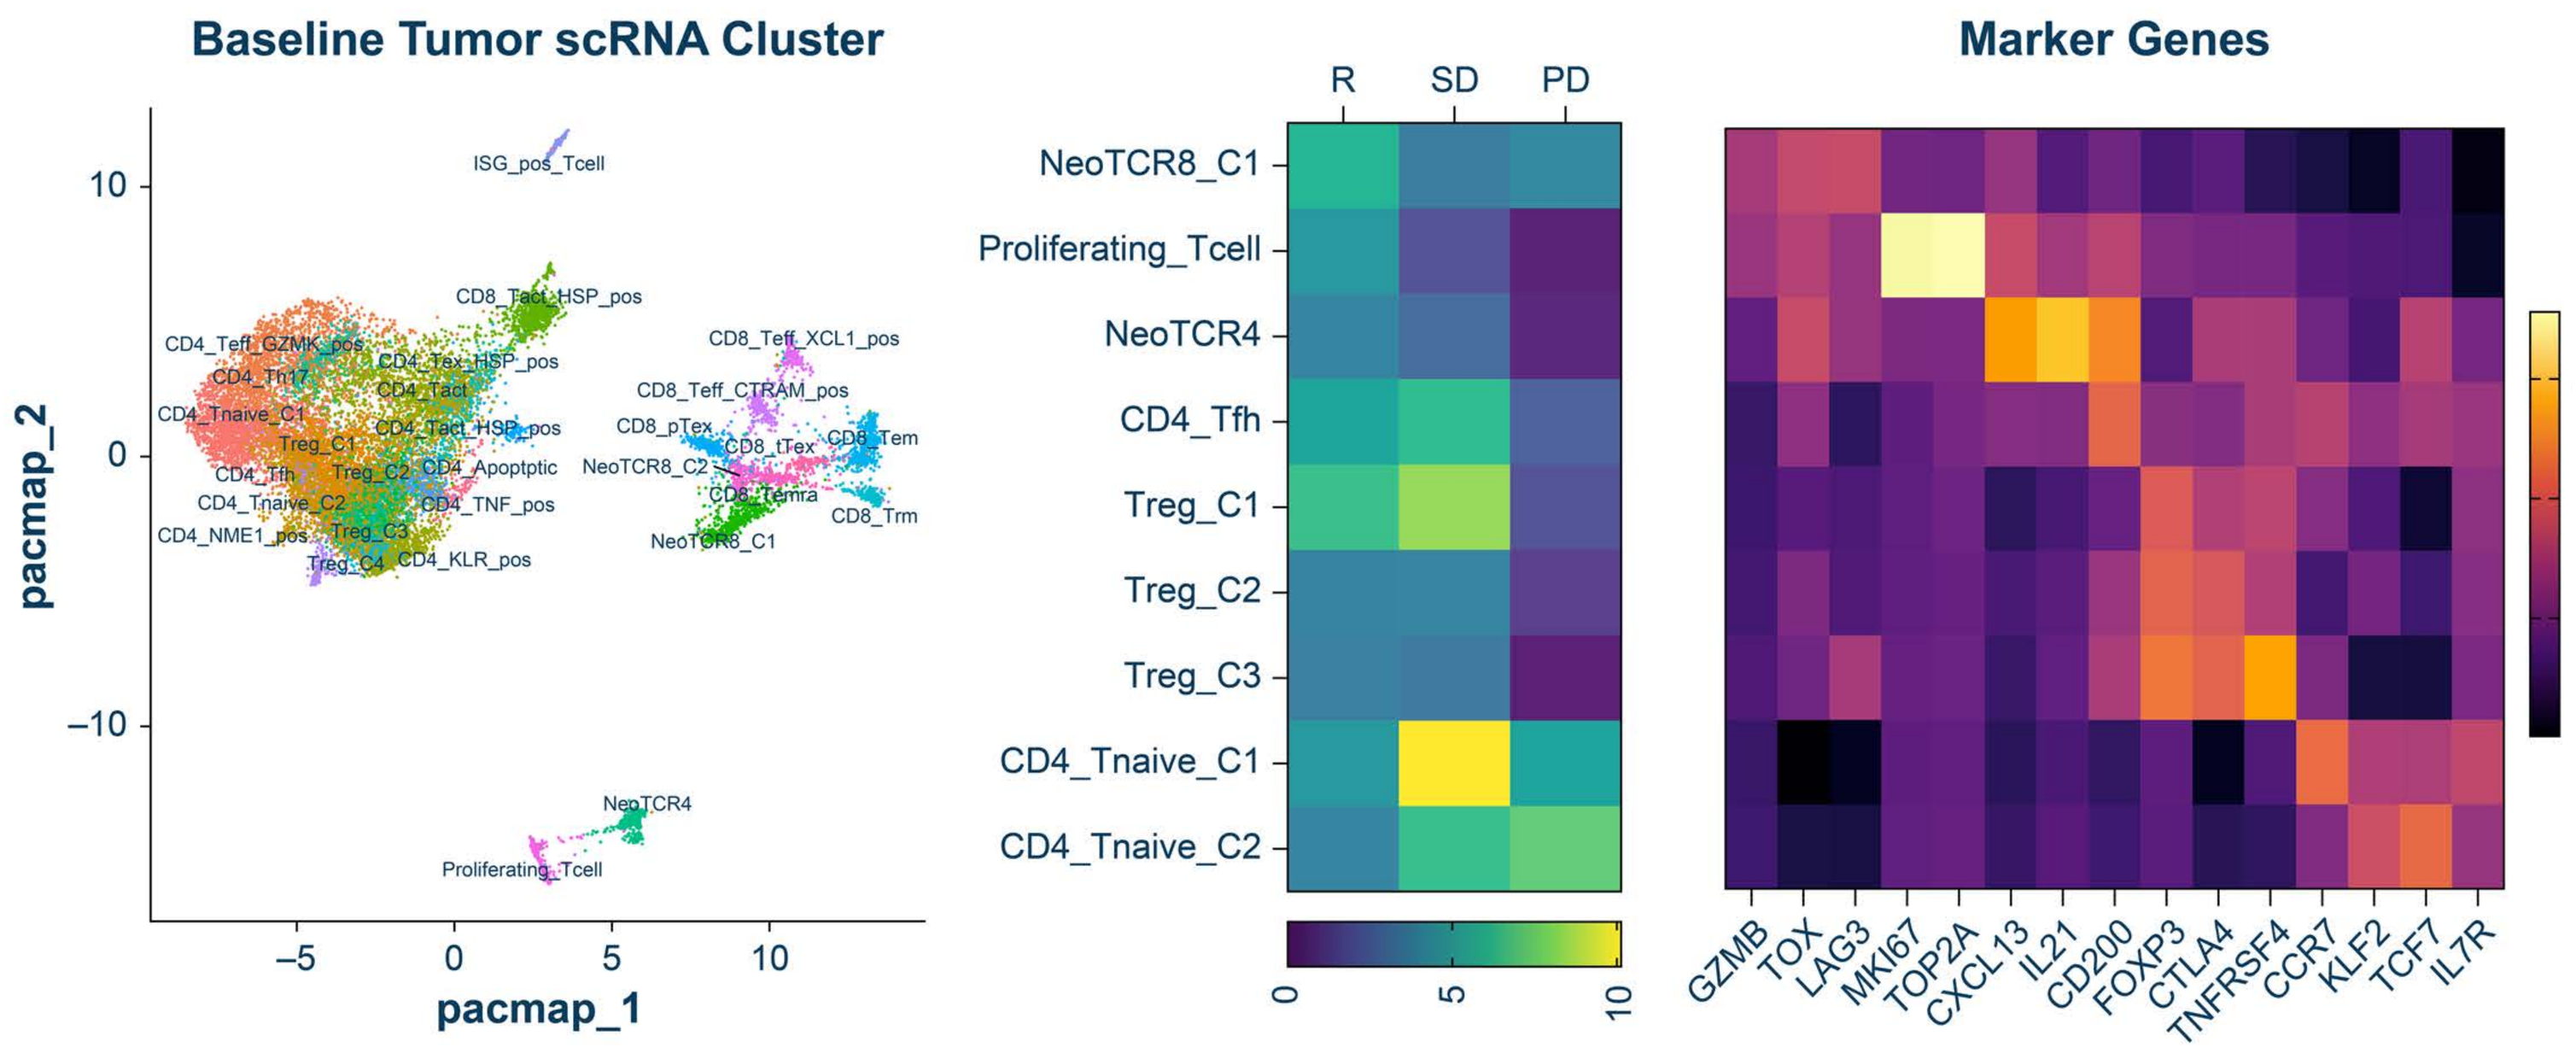
Baseline Tumor scRNA Cluster
pacmap_1
pacmap_2
R
SD
PD
NeoTCR8_C1
Proliferating_Tcell
NeoTCR4
CD4_Tfh
Treg_C1
Treg_C2
Treg_C3
CD4_Tnaive_C1
CD4_Tnaive_C2
Marker Genes
GZMB TOX LAG3 MKI67 TOP2A CXCL13 IL21 CD200 FOXP3 CTLA4 TNFRSF4 CCR7 KLF2 TCF7 IL7R

Figure 3. Profiles of scTCR Clones

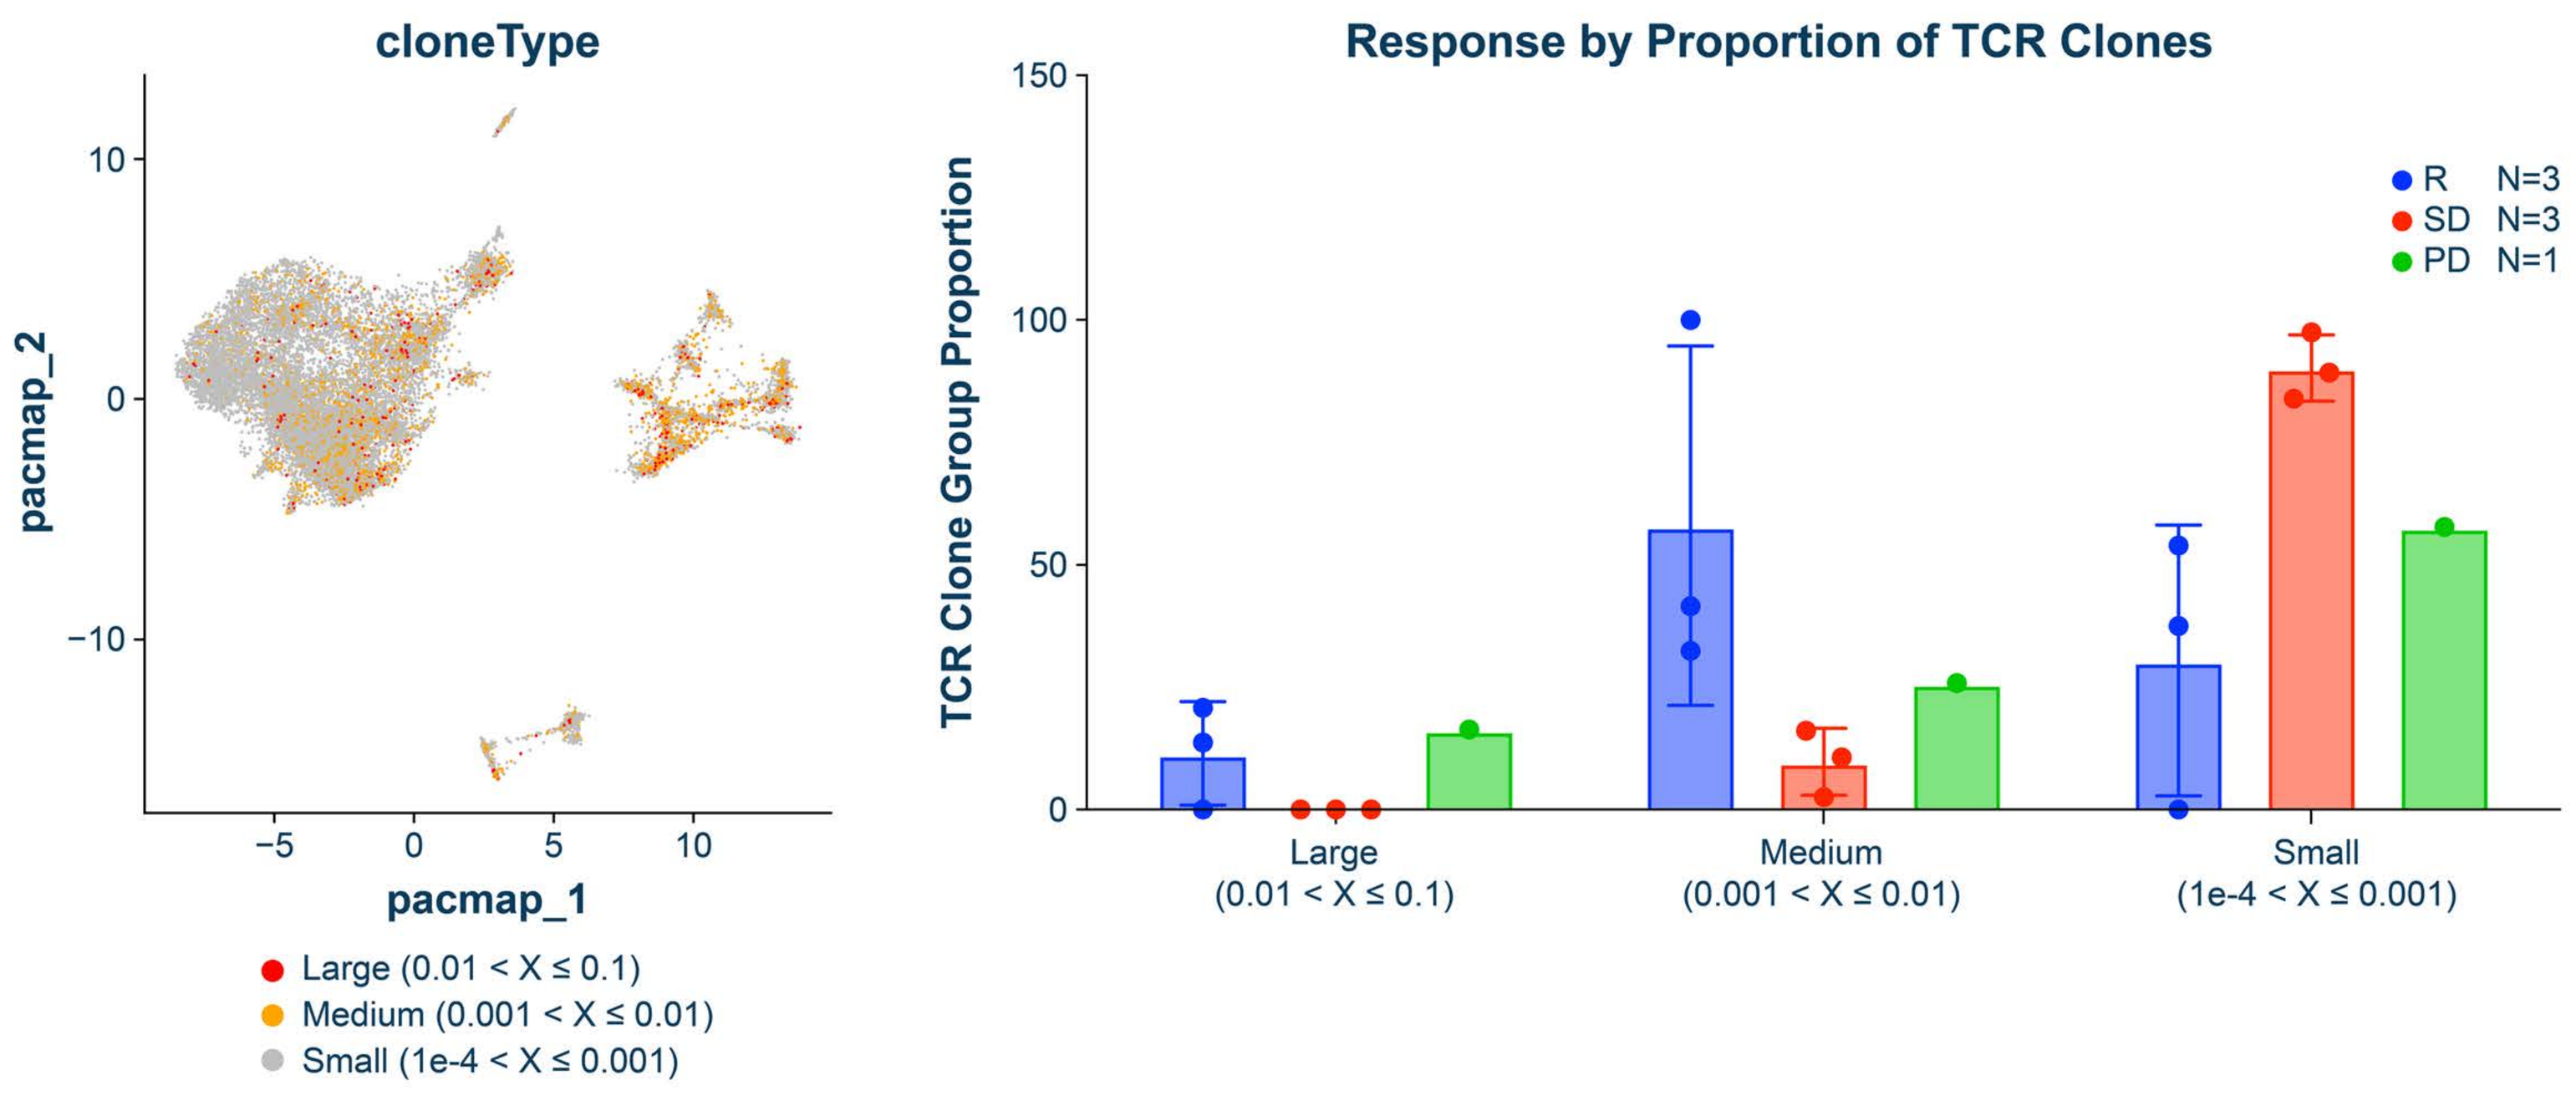
cloneType
pacmap_1
pacmap_2
Large (0.01 < X ≤ 0.1)
Medium (0.001 < X ≤ 0.01)
Small (1e-4 < X ≤ 0.001)
Response by Proportion of TCR Clones
TCR Clone Group Proportion
R N=3
SD N=3
PD N=1
Large (0.01 < X ≤ 0.1)
Medium (0.001 < X ≤ 0.01)
Small (1e-4 < X ≤ 0.001)

Figure 4. NeoTCR T-Cell Proportion in Lifileucel Appears to be Associated with Response

- The TIL DP received by the patient with best overall response of PD lacks this putative tumor-reactive T-cell population (**Figure 4**)
- NeoTCR8 T cells map to CD8 effector memory T-cell (T_{EM})-like cluster in TIL DP
- The lifileucel manufacturing process shifts phenotype of NeoTCR8 T cells
 – NeoTCR8 T cells expand into the billions in lifileucel
 – Upon expansion, NeoTCR8 T cells downregulate exhaustion and upregulate proliferation

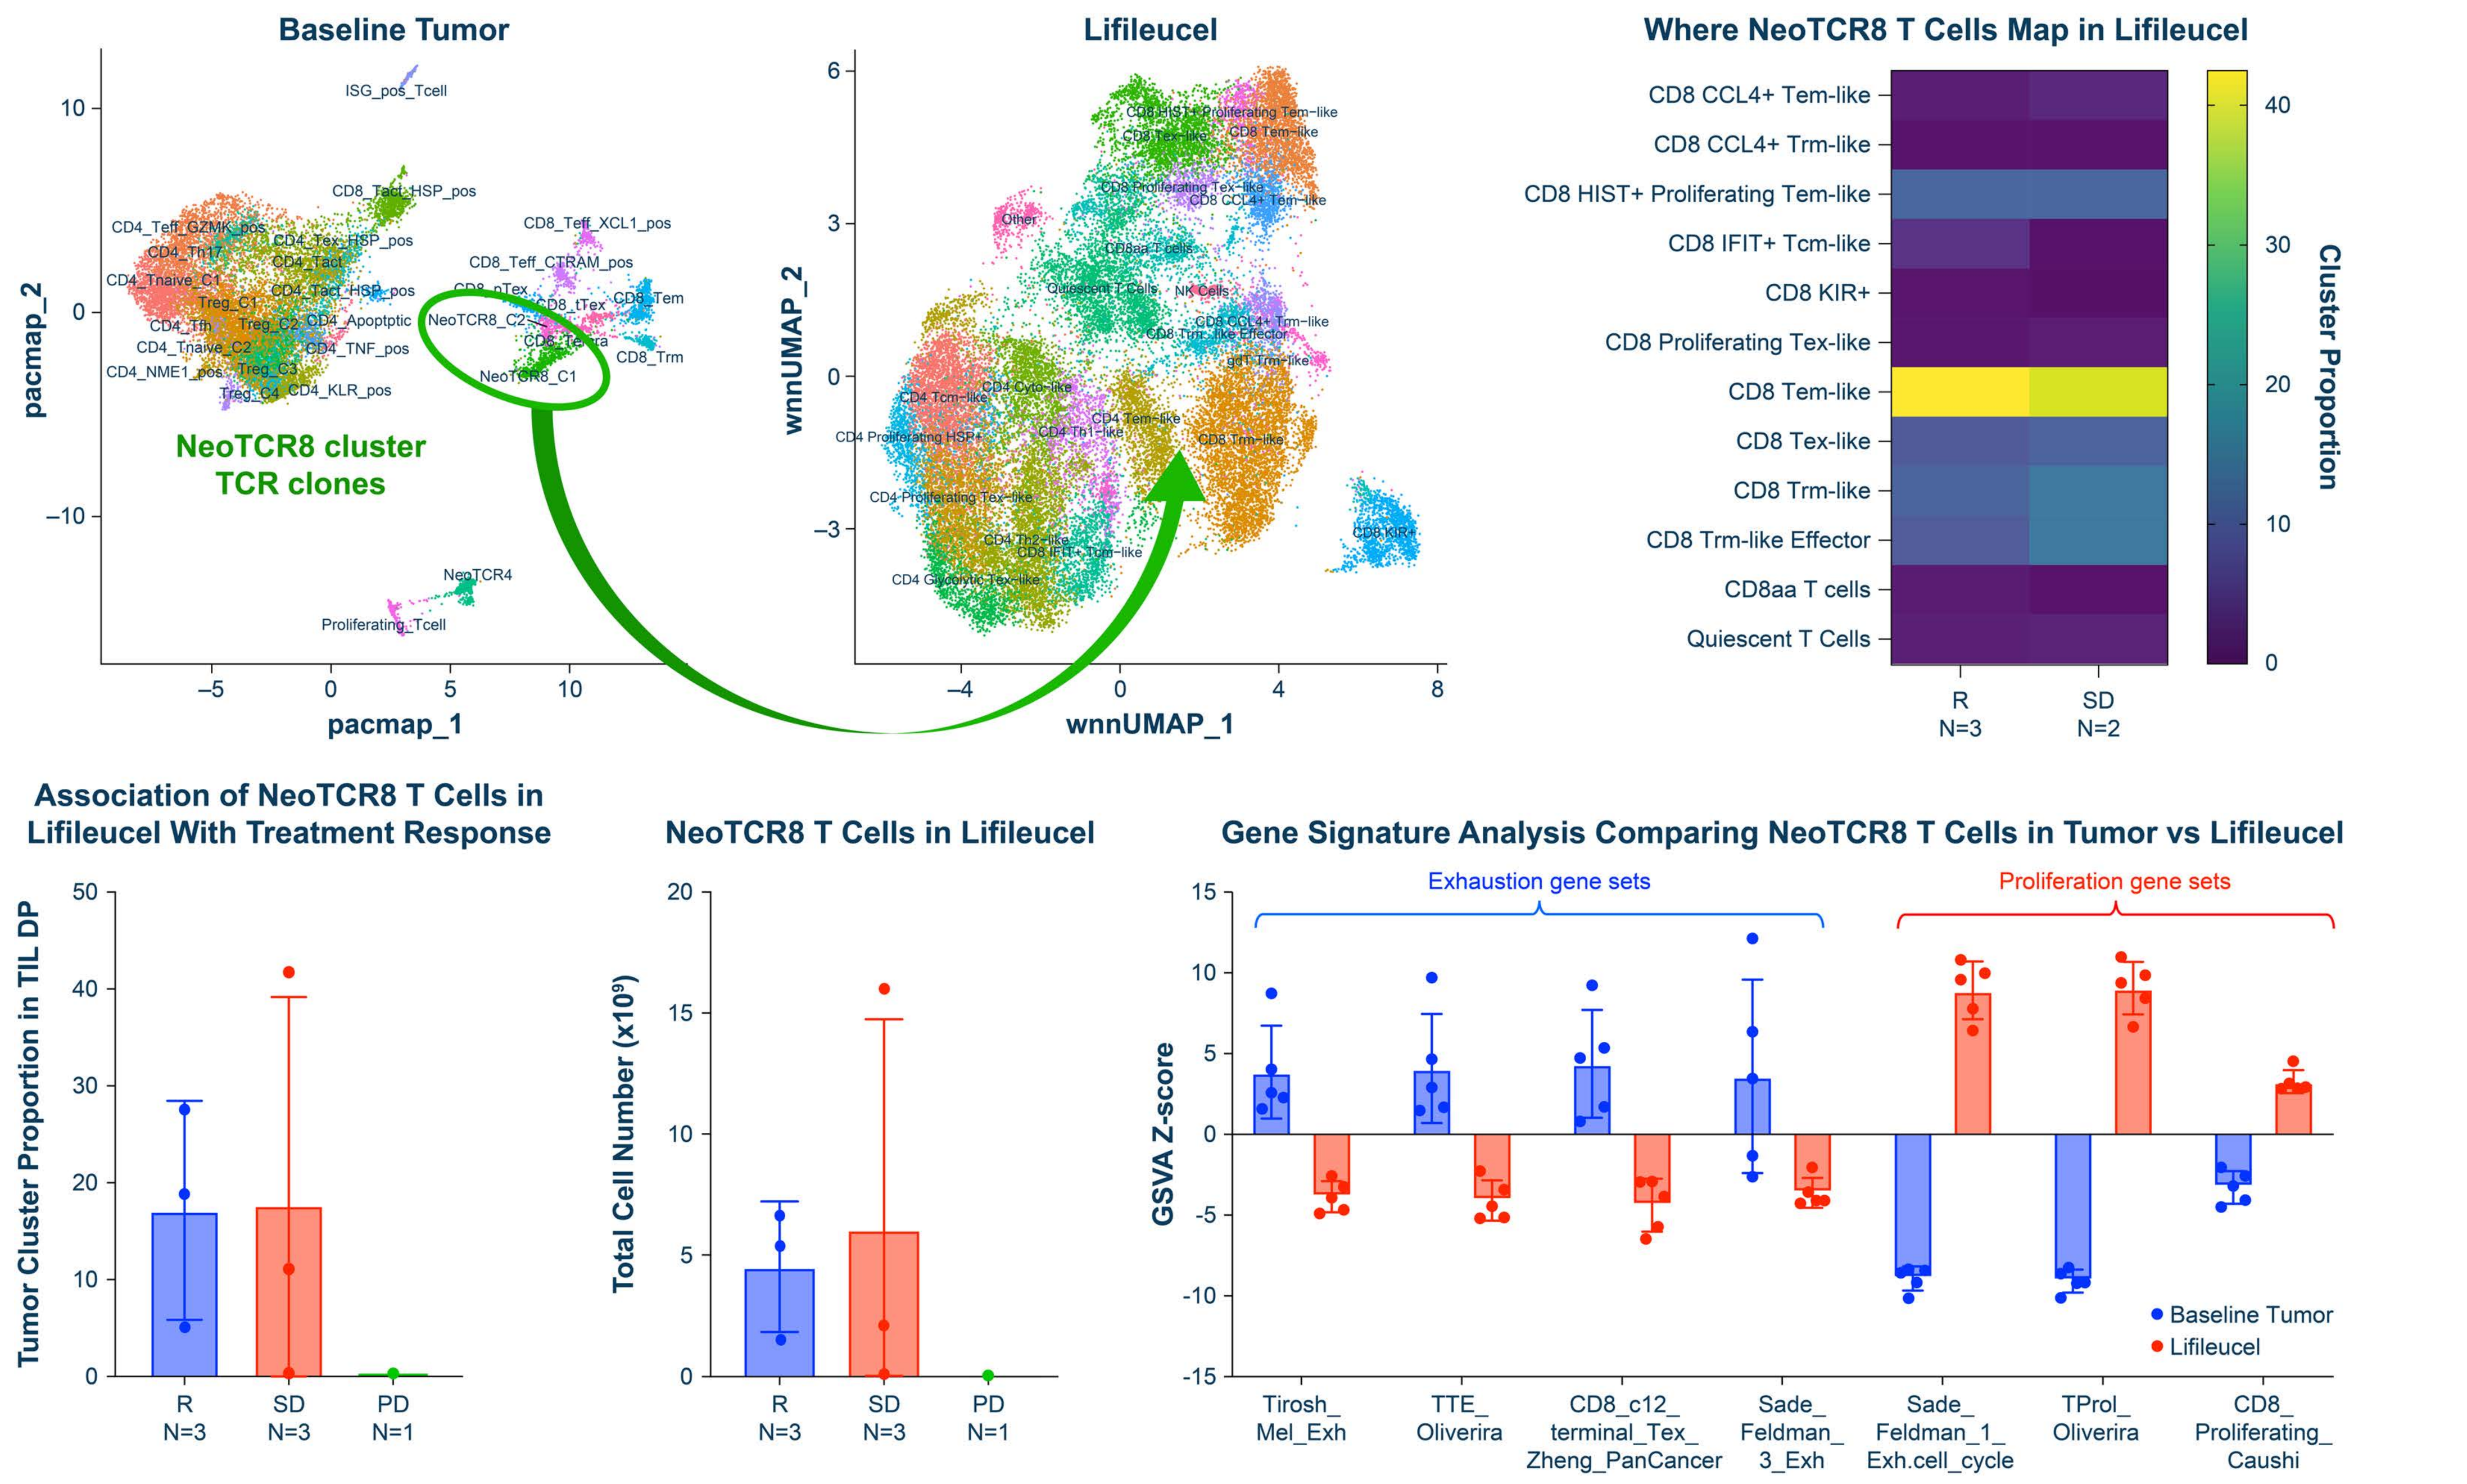
Baseline Tumor
NeoTCR8 cluster TCR clones
pacmap_1
pacmap_2
Lifileucel
wnnUMAP_1
wnnUMAP_2
Where NeoTCR8 T Cells Map in Lifileucel
CD8 CCL4+ Tem-like
CD8 CCL4+ Trm-like
CD8 HIST+ Proliferating Tem-like
CD8 IFIT+ Tcm-like
CD8 KIR+
CD8 Proliferating Tex-like
CD8 Tem-like
CD8 Tex-like
CD8 Trm-like
CD8 Trm-like Effector
CD8aa T cells
Quiescent T Cells
R N=3
SD N=2
Cluster Proportion
Association of NeoTCR8 T Cells in Lifileucel With Treatment Response
Tumor Cluster Proportion in TIL DP
NeoTCR8 T Cells in Lifileucel
Total Cell Number (x10⁹)
R N=3
SD N=3
PD N=1
Gene Signature Analysis Comparing NeoTCR8 T Cells in Tumor vs Lifileucel
Exhaustion gene sets
Proliferation gene sets
GSVA Z-score
Baseline Tumor
Lifileucel
Tirosh_ Mel_Exh
TTE_ Oliverira
CD8_c12_ terminal_Tex_ Zheng_PanCancer
Sade_ Feldman_ 3_Exh
Sade_ Feldman_1_ Exh.cell_cycle
TProl_ Oliverira
CD8_ Proliferating_ Caushi

Figure 5. Single-cell Characterization of Lifileucel Identifies Specific CD8+ TIL Subsets Associated with Response

- The proportion and number of CD8 T_{EM}-like cells in lifileucel is significantly higher in responders in this limited data set (**Figure 5**)
- While generalized associations are observed, the heterogeneity of cell subsets within treatment response groups is clear

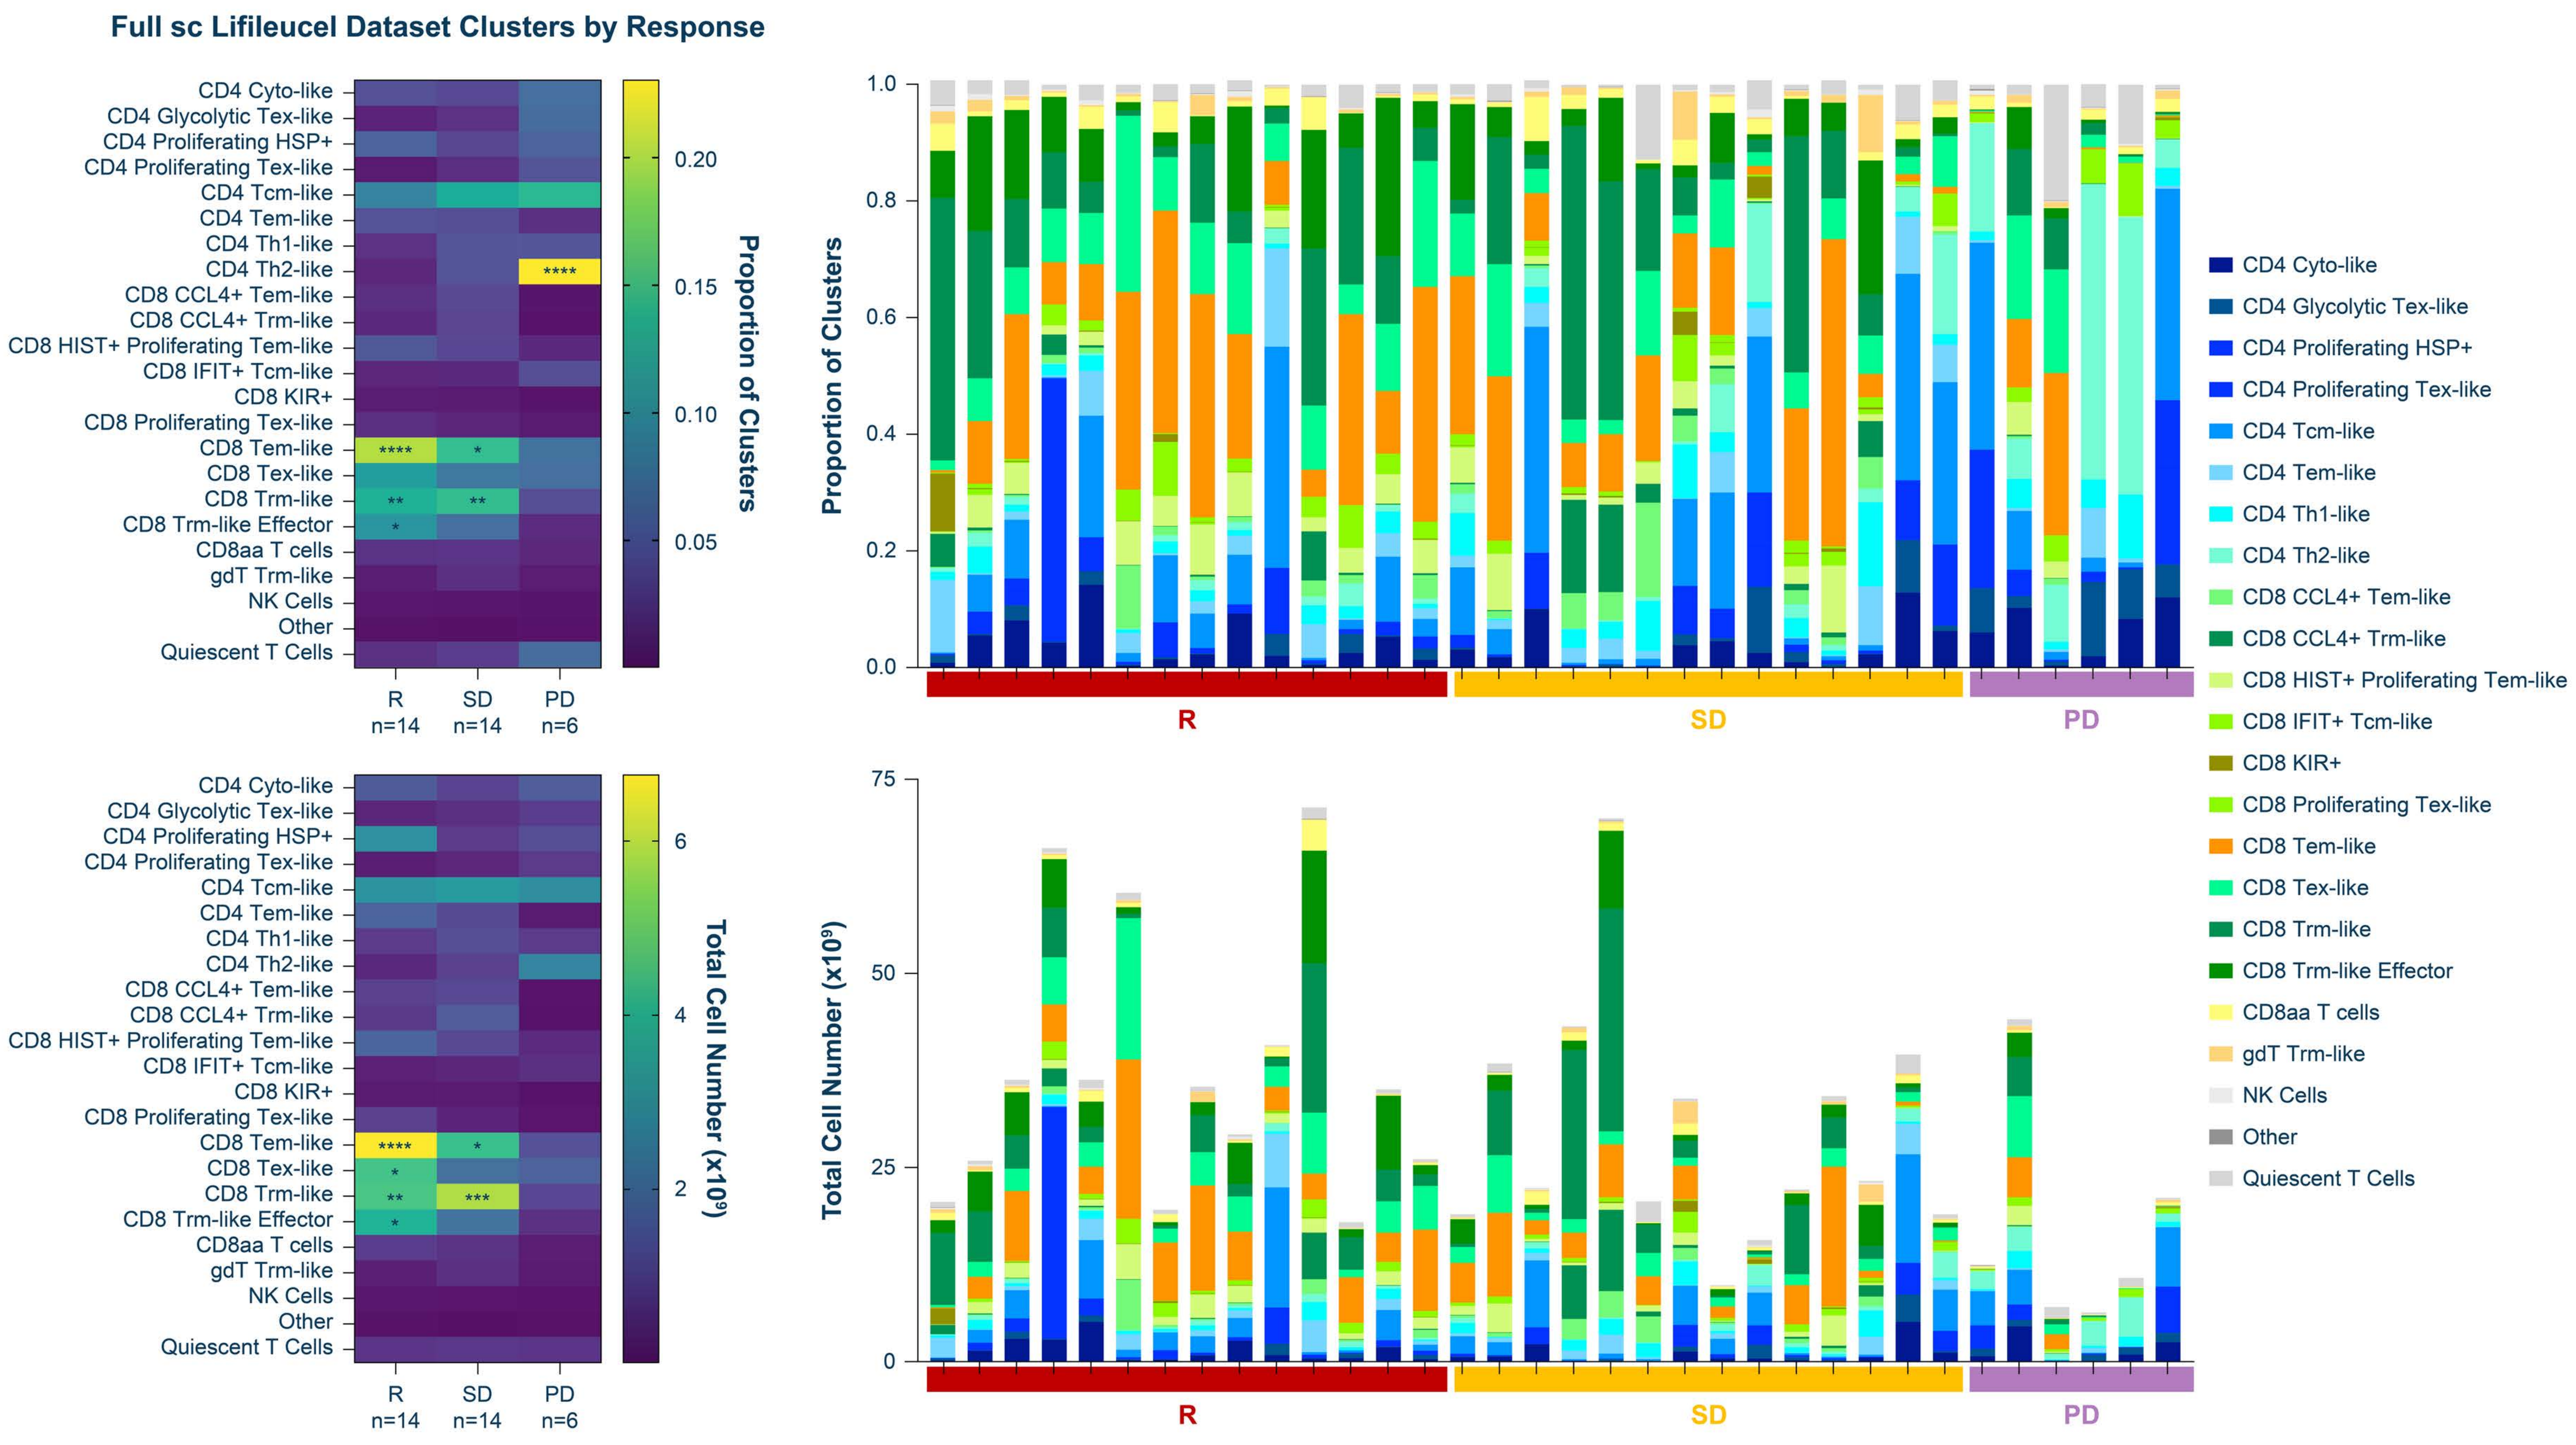
Full sc Lifileucel Dataset Clusters by Response
Proportion of Clusters
Total Cell Number (x10⁹)
R n=14
SD n=14
PD n=6
CD4 Cyto-like
CD4 Glycolytic Tex-like
CD4 Proliferating HSP+
CD4 Proliferating Tex-like
CD4 Tcm-like
CD4 Tem-like
CD4 Th1-like
CD4 Th2-like
CD8 CCL4+ Tem-like
CD8 CCL4+ Trm-like
CD8 HIST+ Proliferating Tem-like
CD8 IFIT+ Tcm-like
CD8 KIR+
CD8 Proliferating Tex-like
CD8 Tem-like
CD8 Tex-like
CD8 Trm-like
CD8 Trm-like Effector
CD8aa T cells
gdT Trm-like
NK Cells
Other
Quiescent T Cells
R
SD
PD

Conclusions

- **The capture, reinvigoration, and expansion of CD8+ TIL-expressing tumor-reactive TCR clones appear to be important for clinical response to TIL cell therapy**
- **Preliminary results suggest the tumor immune infiltrate from patients with stable disease and progressive disease contains more naive CD4 T cells, while responders have more putative tumor-reactive TIL**
- **The lifileucel manufacturing process shifts the phenotype of putative tumor-reactive TIL from exhausted to proliferating T cells**
- **Putative tumor-reactive TIL, post rapid expansion, map to the CD8 T_{EM}-like cluster in the final DP**
- **The proportion and total number of cells in the CD8 T_{EM}-like cluster appears to be associated with response to TIL cell therapy**

References

1. AMTAGVI (lifileucel). https://www.amtagvi.com/. Accessed August 2024.
2. Hao Y, et al. *Nat Biotechnol*. 2024;42:293–304.
3. Hafemeister C, Satija R. *Genome Biol*. 2019;20:296.
4. Korsunsky I, et al. *Nat Methods*. 2019;16:1289–1296.
5. Huang H, et al. *Commun Biol*. 2022;5:719.
6. DeTomaso D, et al. *Nat Commun*. 2019;10:4376.
7. Nofech-Mozes I, et al. *Nat Commun*. 2023;14:1615.
8. Lowery FJ, et al. *Science*. 2022;375(6583):877–84.
9. Borcherding N, et al. *F1000Research*. 2020;9:47.
10. Brunson JC. *J Open Source Softw*. 2017;5:49.
11. Hänzelmann S, et al. *BMC Bioinformatics*. 2013;14:7.

Abbreviations

CITE-seq, cellular indexing of transcriptomes and epitopes by sequencing; DP, drug product; FDA, Food and Drug Administration; GMP, Good Manufacturing Practice; IL-2, interleukin-2; PD, progressive disease; R, responder; scRNA, single-cell RNA; scTCR, single-cell T-cell receptor; SD, stable disease; T_{CM}, central memory T cell; TCR, T-cell receptor; T_{EM}, effector memory T cell; Tex, exhausted T cell; TIL, tumor-infiltrating lymphocyte; Treg, regulatory T cell; T_{RM}, tissue resident memory T cell.

Acknowledgments

• This study is sponsored by Iovance Biotherapeutics, Inc. (San Carlos, CA, USA)
• Medical writing and editorial support was provided by Peloton Advantage, LLC, an OPEN Health company, and funded by Iovance
• Special thank you to the C-144-01 study patients and their families

Disclosures

Theresa Medina: BioAtla, Bristol Myers Squibb, Checkmate, Day One Pharmaceutical, Exicure, Iovance Biotherapeutics, Merck, Moderna, Nektar, Pfizer, Regeneron, Replimune, Taiga, Xencor; **Amod Sarnaik:** Blueprint Oncology Concepts, Gerson Lehrman Group, Guidepoint, Iovance Biotherapeutics, Provectus Biopharmaceuticals, Second City Science, Turnstone Biologics; **Jason Chesney:** Amgen, Iovance Biotherapeutics, Obsidian, Replimune; **Mike Cusnir:** None to disclose; **Joe Dean, Joe Yglesias, Behzad Damirchi, Mark Ozeck, Kranthi Kunkalla, Brian Gastman, Rana Fiaz, Giri Sulur, Hequn Yin, Rongsu Qi:** Iovance Biotherapeutics.

© 2025, Iovance Biotherapeutics